Eblasakimab Significantly Alleviates IL-4 and IL-13-Induced Bronchial Airway Constriction in COPD-Derived Lung Slices

Ferda Cevikbas,[1] Paul E Miller,[2] and Carl Firth[1] [1]ASLAN Pharmaceuticals, San Mateo, CA, USA and Singapore; [2]Mechanobiologix, LLC, Newton, MA, USA

INTRODUCTION

- Patients with chronic obstructive pulmonary disorder (COPD) are vulnerable to acute exacerbations characterized by impaired airflow, obstruction, and dyspnea, underscoring the need for preventing exacerbations[1,2]
- Growing evidence indicates that Type 2 inflammation is observed during COPD exacerbations as well as during stable disease states in some patients[3]
 - In a Phase 3, double blind, randomized trial, COPD patients with Type 2 inflammation (indicated by elevated blood eosinophil counts) and a higher exacerbation risk treated with dupilumab had fewer exacerbations at week 52 compared to placebo-treated patients[4]
- Since interleukin-4 (IL-4) and interleukin-13 (IL-13) are central mediators of Type 2 inflammation,[5] and are implicated in various mechanisms of airway obstruction in asthma,[3] they may play an essential role in COPD[3]
- Therefore, blocking dual IL-4 and IL-13 signaling may have the potential to prevent bronchoconstriction evoked by the dual action of these proinflammatory cytokines
- IL-4 signaling is conducted through both Type I (IL-4Rα and γC) and Type II receptors (IL-4Rα and IL-13Rα1), while IL-13 signals through the Type II receptor[6]
- Dupilumab, an antibody approved for the treatment of multiple indications including atopic dermatitis and asthma, and eblasakimab, an antibody currently in clinical development for atopic dermatitis, are biologics that target different receptor subunits of the IL-4 and IL-13 signaling pathway
- While dupilumab functions by targeting IL-4Rα,[7] shared by both the Type I and II receptors, eblasakimab, a human IgG4 monoclonal antibody, targets the IL-13Rα1 subunit, uniquely blocking signaling through the Type II receptor complex and sparing the Type I receptor with the potential to block the Type II receptor more efficiently[8] (**Figure 1**)

FIGURE 1. EBLASAKIMAB AND DUPILUMAB MECHANISM OF ACTION

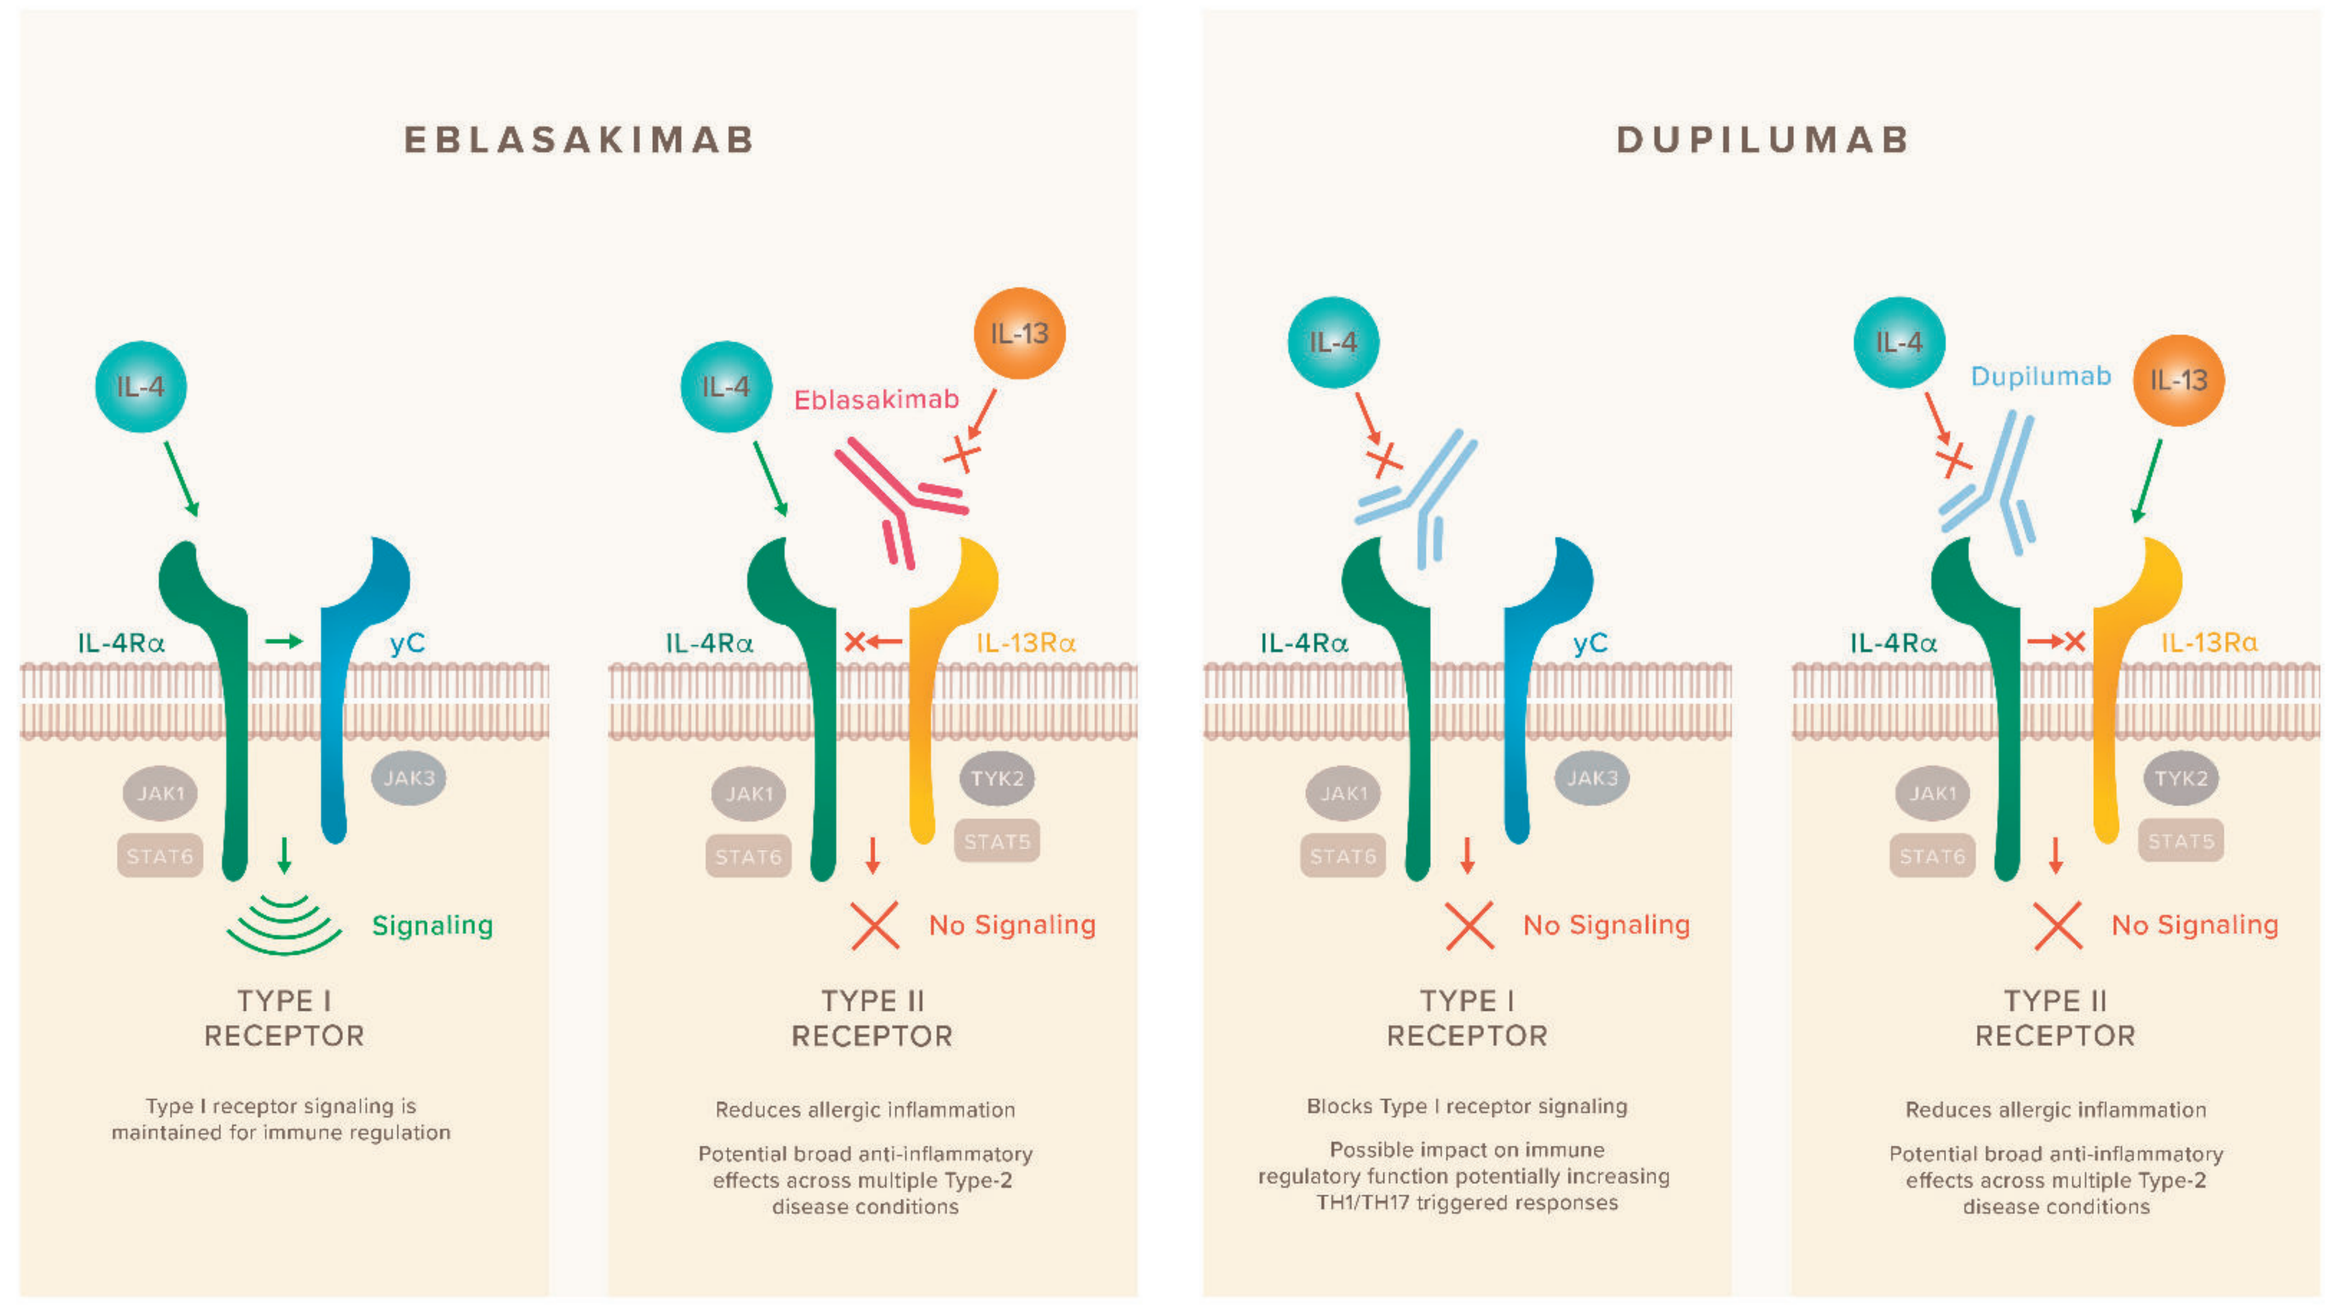
EBLASAKIMAB
DUPILUMAB
IL-4
IL-13
Eblasakimab
Dupilumab
IL-4Rα
yC
IL-13Rα
JAK1
JAK3
TYK2
STAT6
STAT5
Signaling
No Signaling
TYPE I RECEPTOR
TYPE II RECEPTOR
Type I receptor signaling is maintained for immune regulation
Reduces allergic inflammation
Potential broad anti-inflammatory effects across multiple Type-2 disease conditions
Blocks Type I receptor signaling
Possible impact on immune regulatory function potentially increasing TH1/TH17 triggered responses

IL-4, interleukin-4; IL-13, interleukin-13; IL-4Rα, interleukin-4 receptor alpha; IL-13Rα, interleukin-13 receptor alpha 1.

OBJECTIVE

- Investigate the impact of eblasakimab on IL-4- and IL-13-induced bronchial airway constriction and compare the efficacy of eblasakimab with dupilumab in a human ex-vivo translational model for COPD.

KEY RESULTS: PRELIMINARY DATA INDICATE THAT EBLASAKIMAB HAS THE POTENTIAL TO SIGNIFICANTLY IMPROVE IL-4 AND IL-13-INDUCED BRONCHIAL AIRWAY CONSTRICTION

- In healthy PCLS, IL-4 and IL-13 combination treatment induced significantly higher percentages of bronchial airway constriction (p<0.01 vs control; **Figure 3A**), suppressed bronchial dilation levels stimulated by the relaxing agent formoterol (p<0.05 vs control; **Figure 3B**), and enhanced methacholine-induced airway constriction (p<0.05 vs control; **Figure 3C**)
- Both eblasakimab and dupilumab treatment significantly counteracted the constricting impacts of IL-4 and IL-13 cytokine exposure, restoring levels of airway constriction and dilation to levels observed in non-cytokine treated control slices (**Figures 3A-C**)
 - Both high and low doses of eblasakimab restored formoterol-induced acute dilation (5 minutes) in IL-4 and IL-13 pre-treated airways to levels observed in non-cytokine treated control slices (**Figure 3B**)
 - A low dose of eblasakimab was effective at inhibiting methacholine-induced constriction to levels observed in non-cytokine treated control slices (**Figure 3C**)
- In COPD-derived PCLS, exposure to IL-4 and IL-13 induced similar effects as in healthy tissue: significantly higher percentages of bronchial airway constriction (p<0.01 vs control; **Figure 4A**), numerically lower percentages of acute airway dilation in response to formoterol exposure compared to controls (**Figure 4B**), and increased constriction in response to methacholine (p<0.001 vs control; **Figure 4C**)
- Eblasakimab shows significant improvement compared with placebo across all measured bronchial outcomes in COPD-derived precision cut lung slices whereas dupilumab effects did not achieve statistical significance relative to placebo (**Figures 4A-C**)

FIGURE 3. BRONCHIAL AIRWAY RESPONSES TO VARIOUS TREATMENT CONDITIONS IN HEALTHY HUMAN PRECISION CUT LUNG SLICES

A. Percent Bronchial Contraction in Response to Different Treatment Conditions

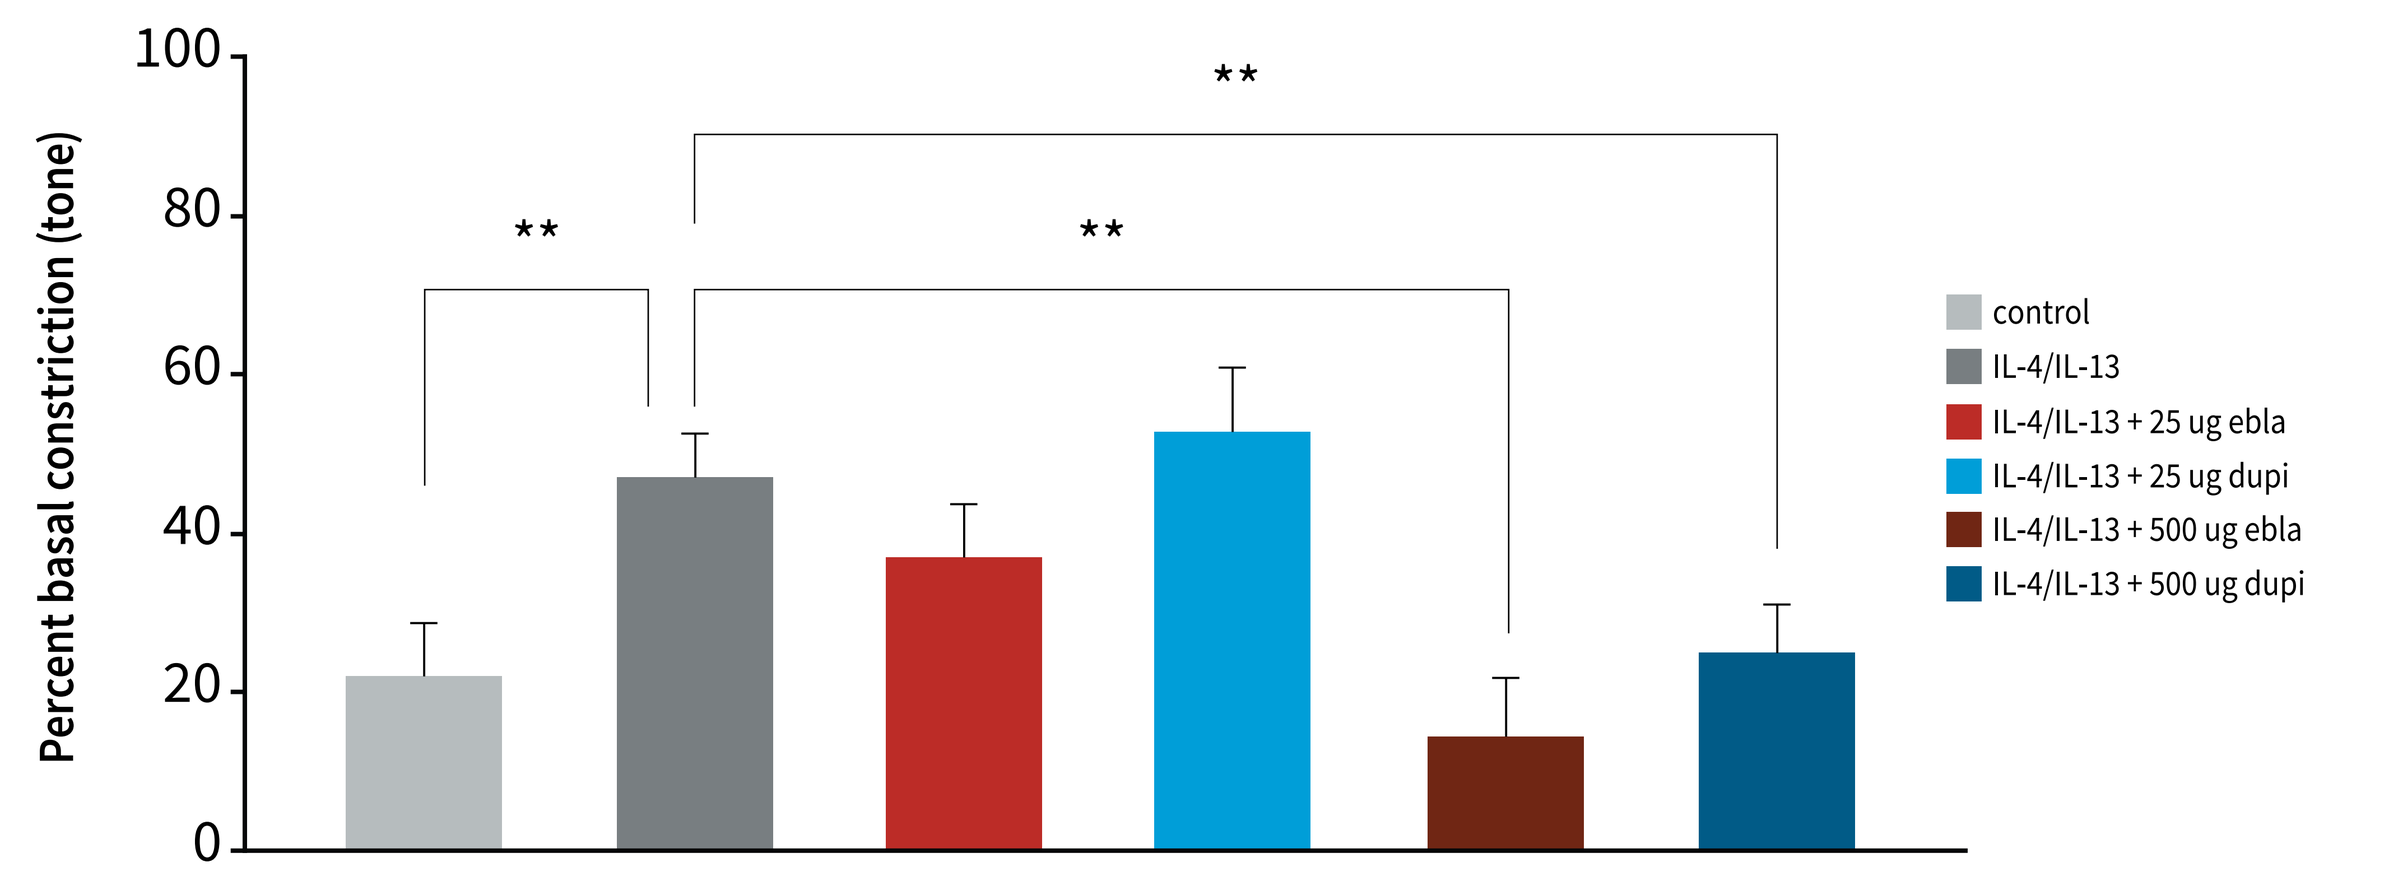
Percent basal constriction (tone)
100
80
60
40
20
0
**
**
**
control
IL-4/IL-13
IL-4/IL-13 + 25 ug ebla
IL-4/IL-13 + 25 ug dupi
IL-4/IL-13 + 500 ug ebla
IL-4/IL-13 + 500 ug dupi

B. Percent Bronchial Dilation in Response to Different Treatment Conditions and Subsequent Formoterol Exposure

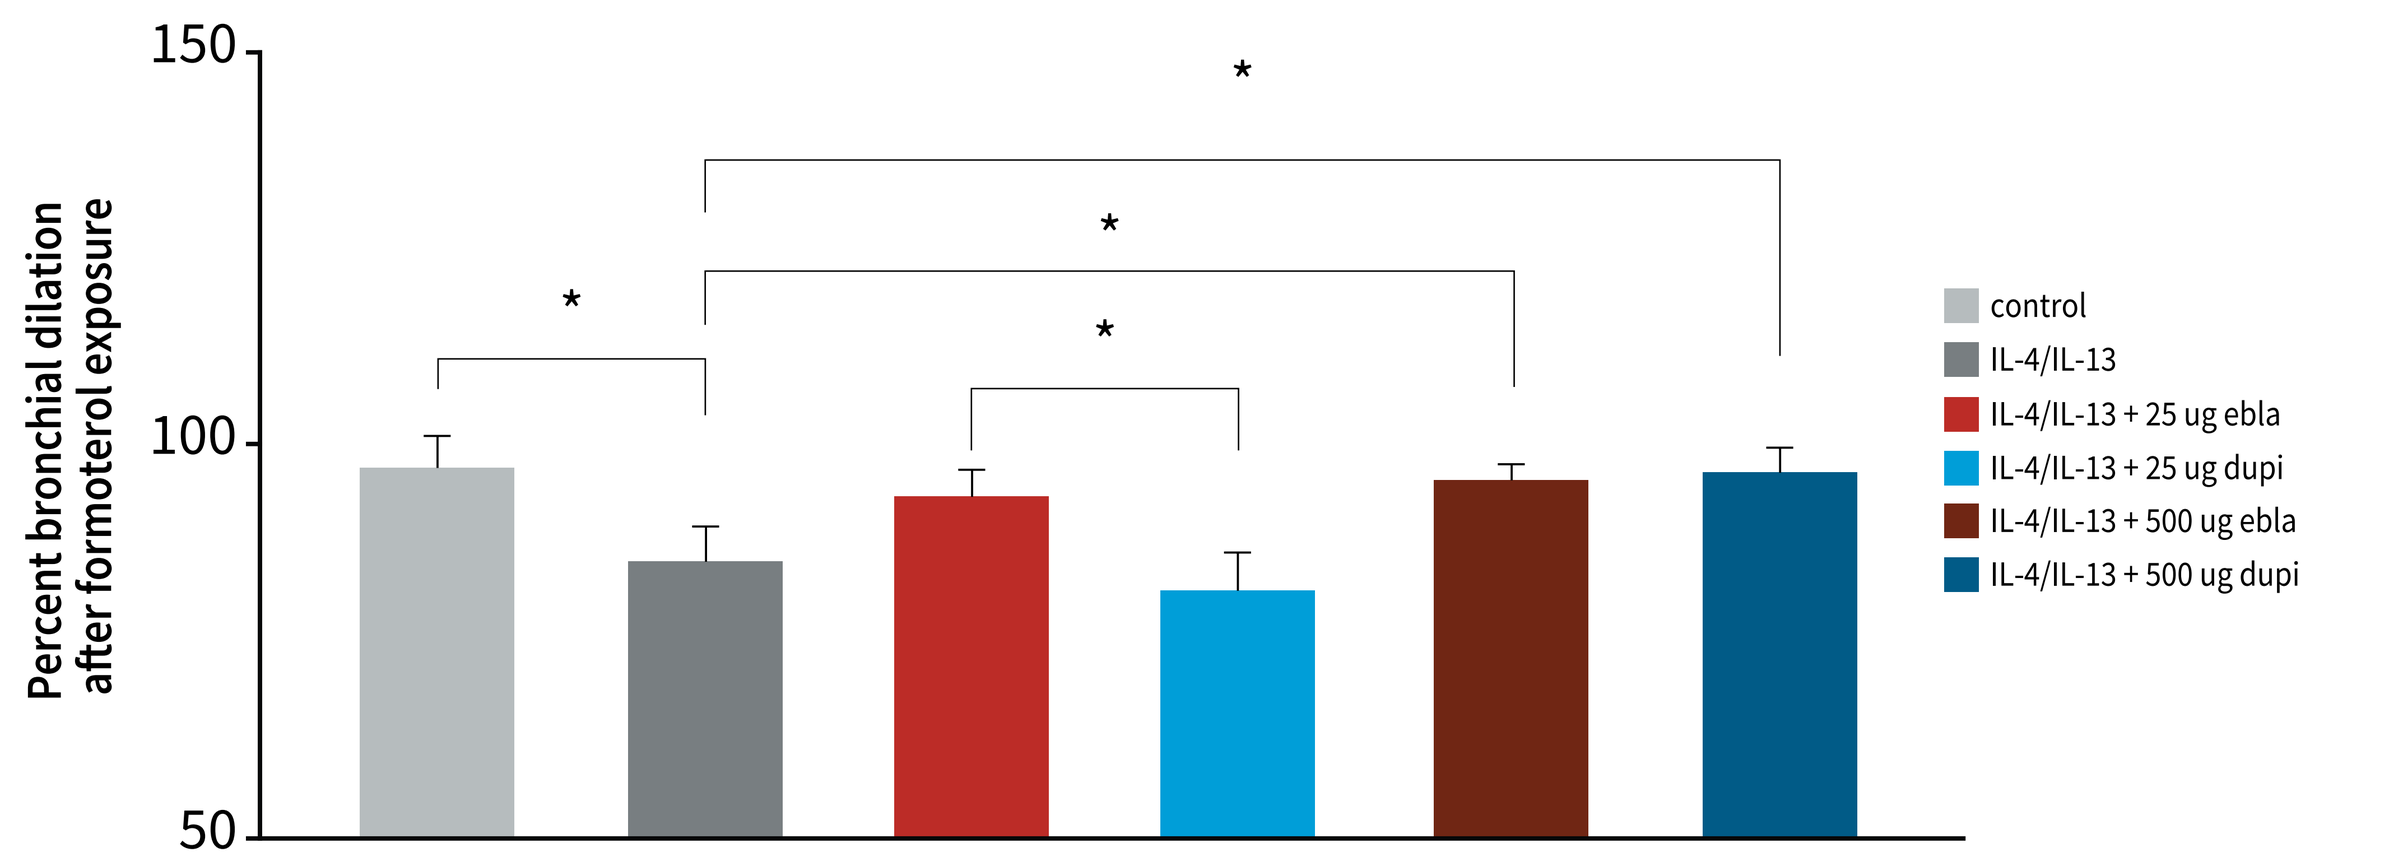
Percent bronchial dilation after formoterol exposure
150
100
50
*
*
*
*
control
IL-4/IL-13
IL-4/IL-13 + 25 ug ebla
IL-4/IL-13 + 25 ug dupi
IL-4/IL-13 + 500 ug ebla
IL-4/IL-13 + 500 ug dupi

C. Percent Bronchial Contraction in Response to Different Treatment Conditions and Subsequent Methacholine Exposure

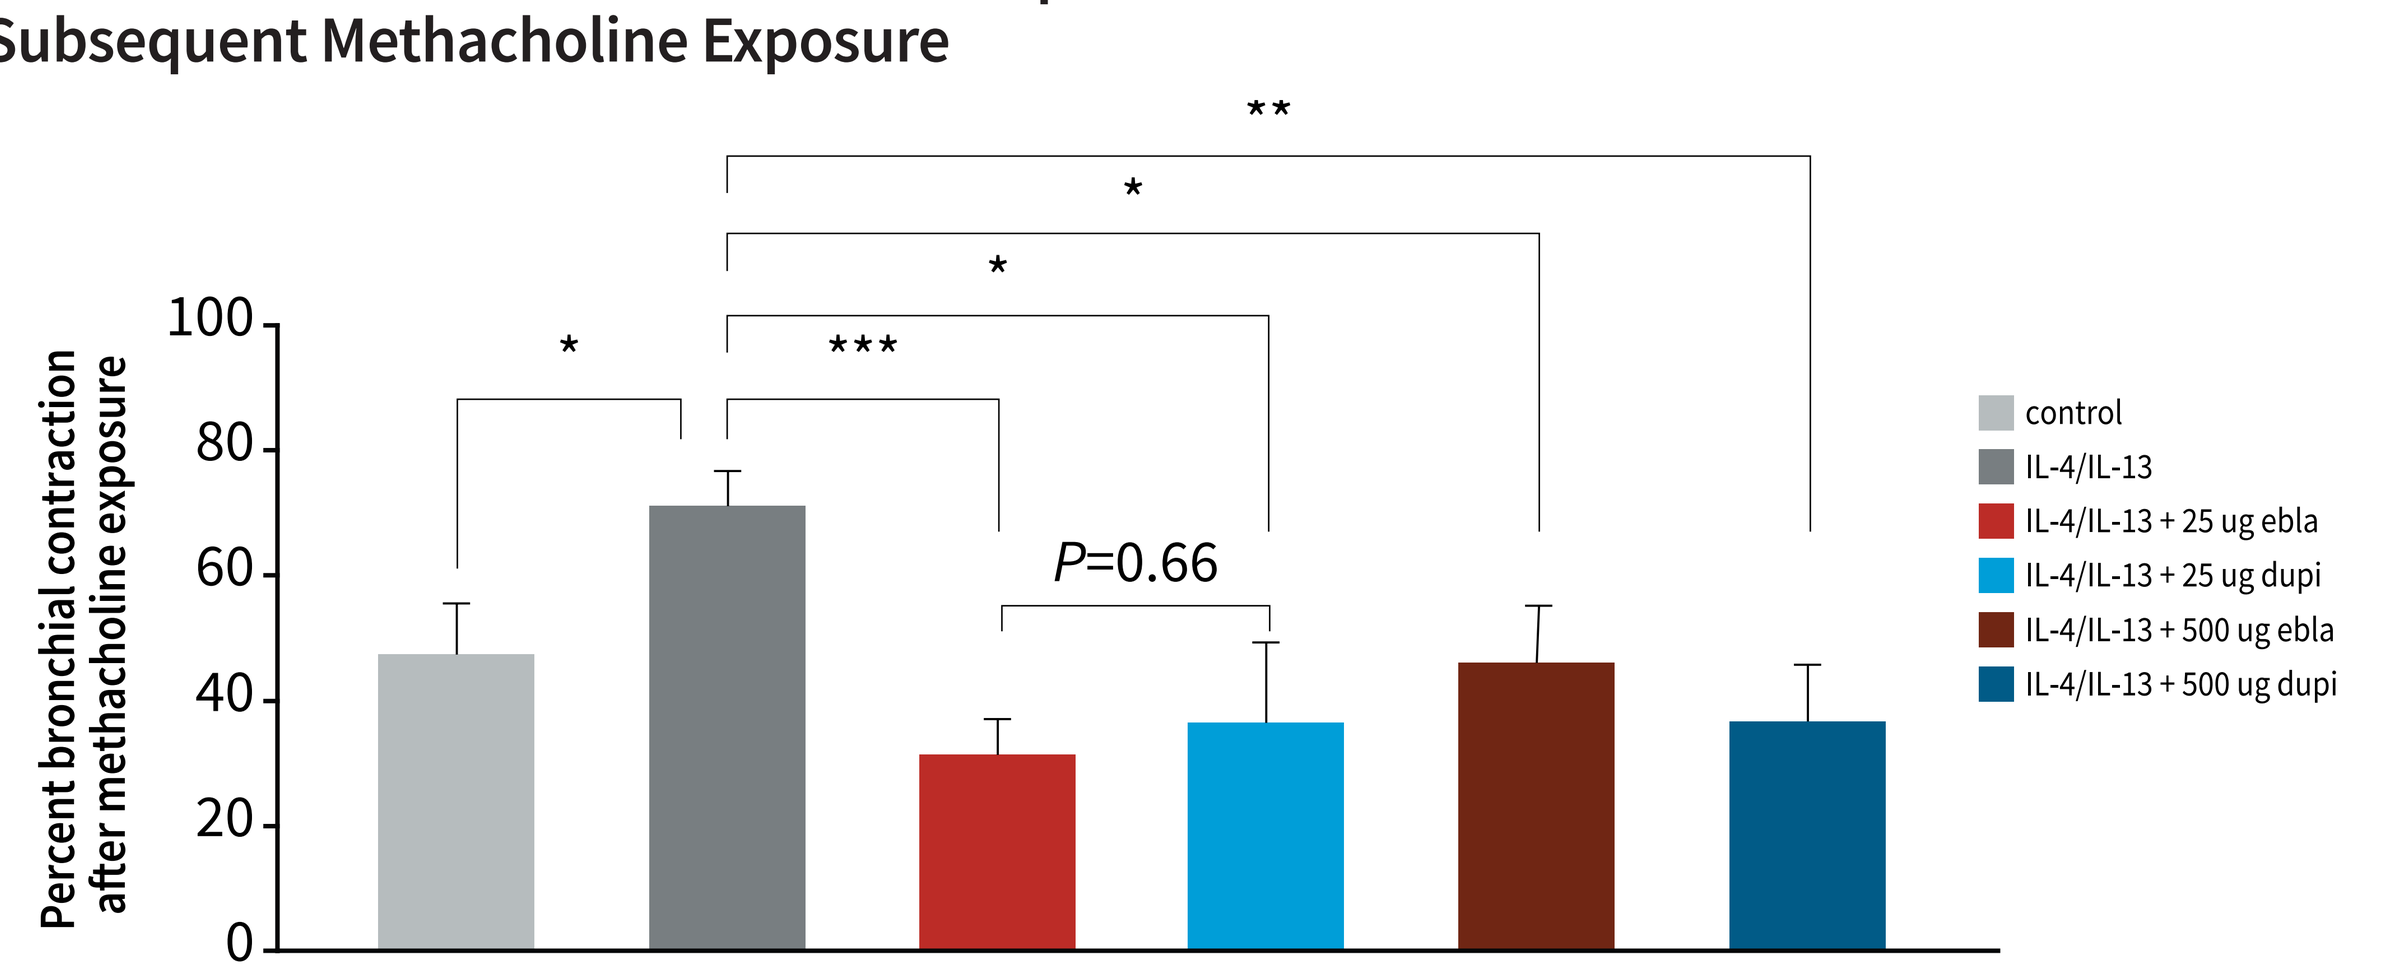
Percent bronchial contraction after methacholine exposure
100
80
60
40
20
0
**
*
*
*

P=0.66
control
IL-4/IL-13
IL-4/IL-13 + 25 ug ebla
IL-4/IL-13 + 25 ug dupi
IL-4/IL-13 + 500 ug ebla
IL-4/IL-13 + 500 ug dupi

Dupi, dupilumab; ebla, eblasakimab; IL-4, interleukin-4; IL-13, interleukin-13.
n=6-13 airways per condition.
p*<0.05; *p*<0.01; ****p*<0.001.

FIGURE 4. BRONCHIAL AIRWAY RESPONSES TO VARIOUS TREATMENT CONDITIONS IN COPD-DERIVED PRECISION CUT LUNG SLICES

A. Percent Bronchial Contraction in Response to Different Treatment Conditions

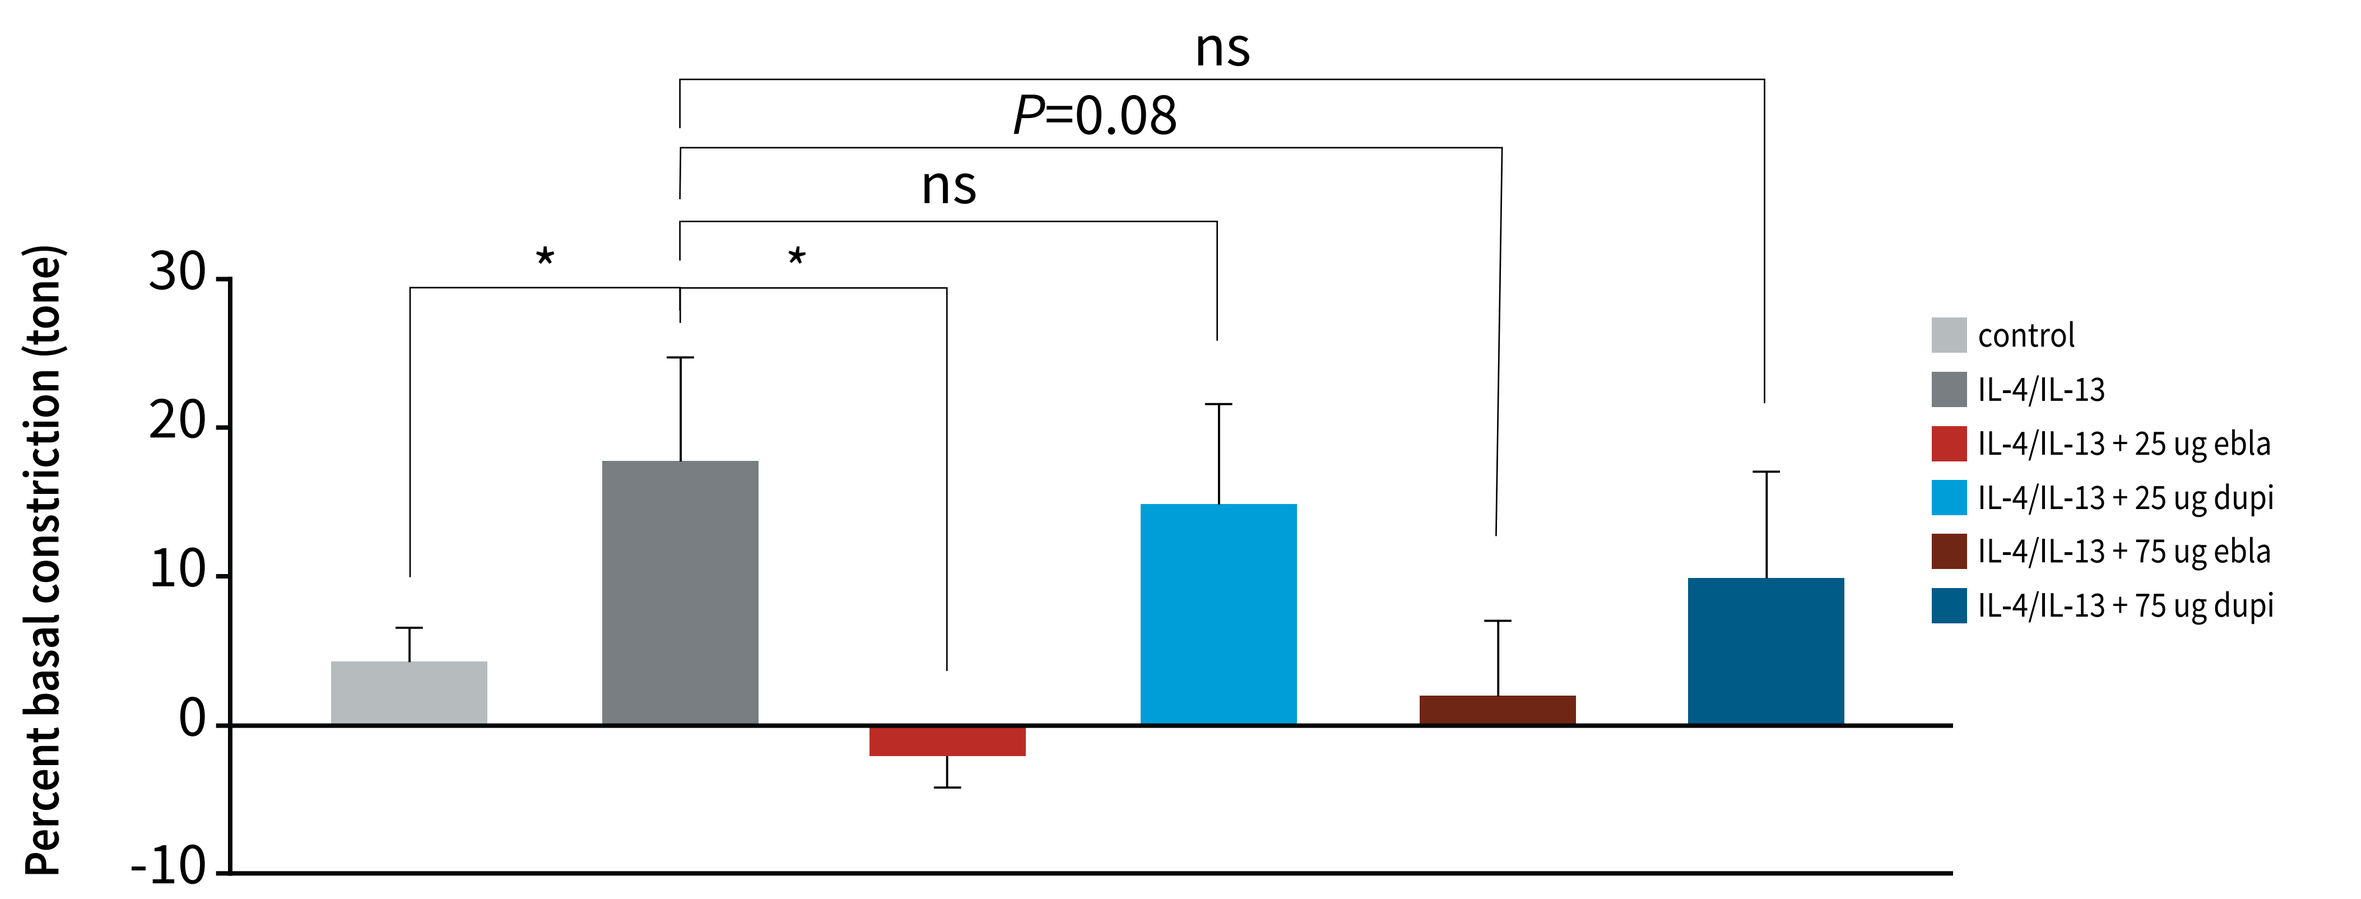
Percent basal constriction (tone)
30
20
10
0
-10
ns
P=0.08
ns
*
*
control
IL-4/IL-13
IL-4/IL-13 + 25 ug ebla
IL-4/IL-13 + 25 ug dupi
IL-4/IL-13 + 75 ug ebla
IL-4/IL-13 + 75 ug dupi

B. Percent Bronchial Dilation in Response to Different Treatment Conditions and Subsequent Formoterol Exposure

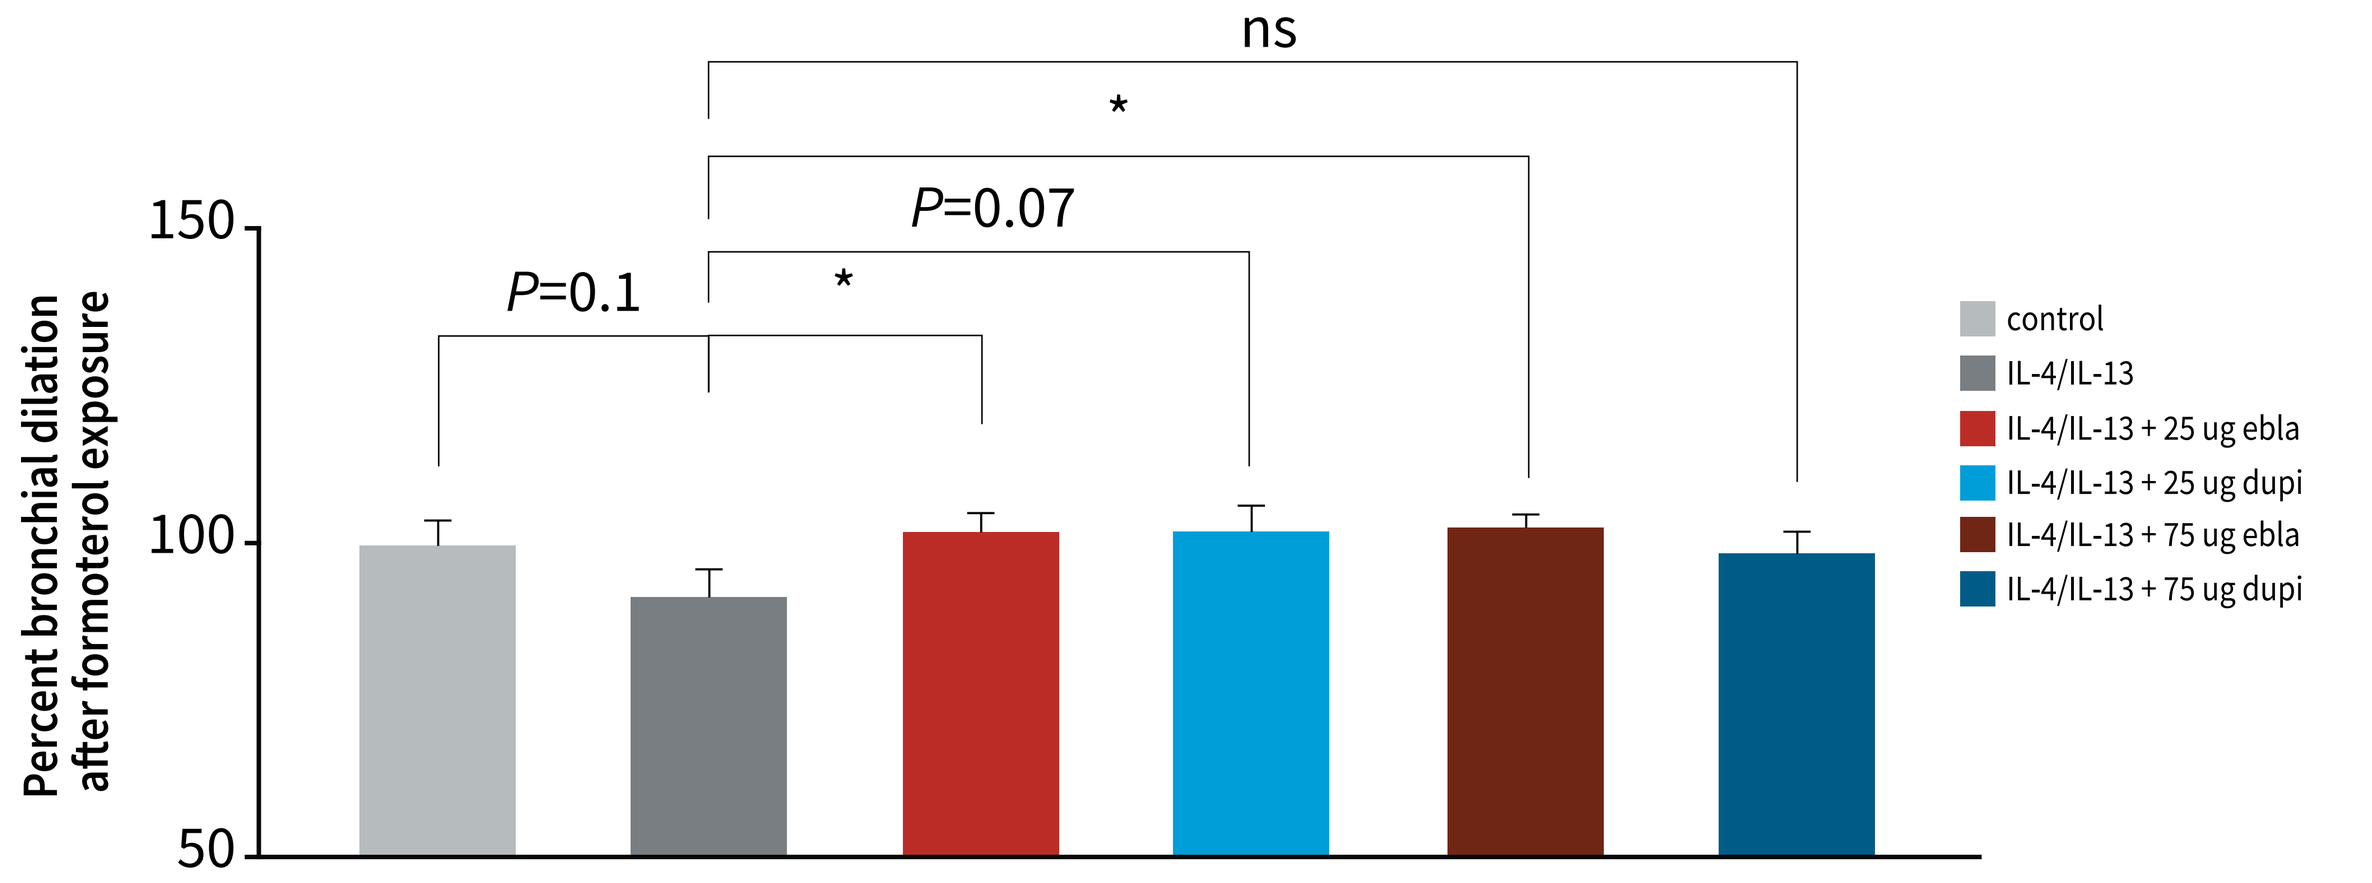
Percent bronchial dilation after formoterol exposure
150
100
50
ns
*
P=0.07
P=0.1
*
control
IL-4/IL-13
IL-4/IL-13 + 25 ug ebla
IL-4/IL-13 + 25 ug dupi
IL-4/IL-13 + 75 ug ebla
IL-4/IL-13 + 75 ug dupi

C. Percent Bronchial Contraction in Response to Different Treatment Conditions and Subsequent Methacholine Exposure

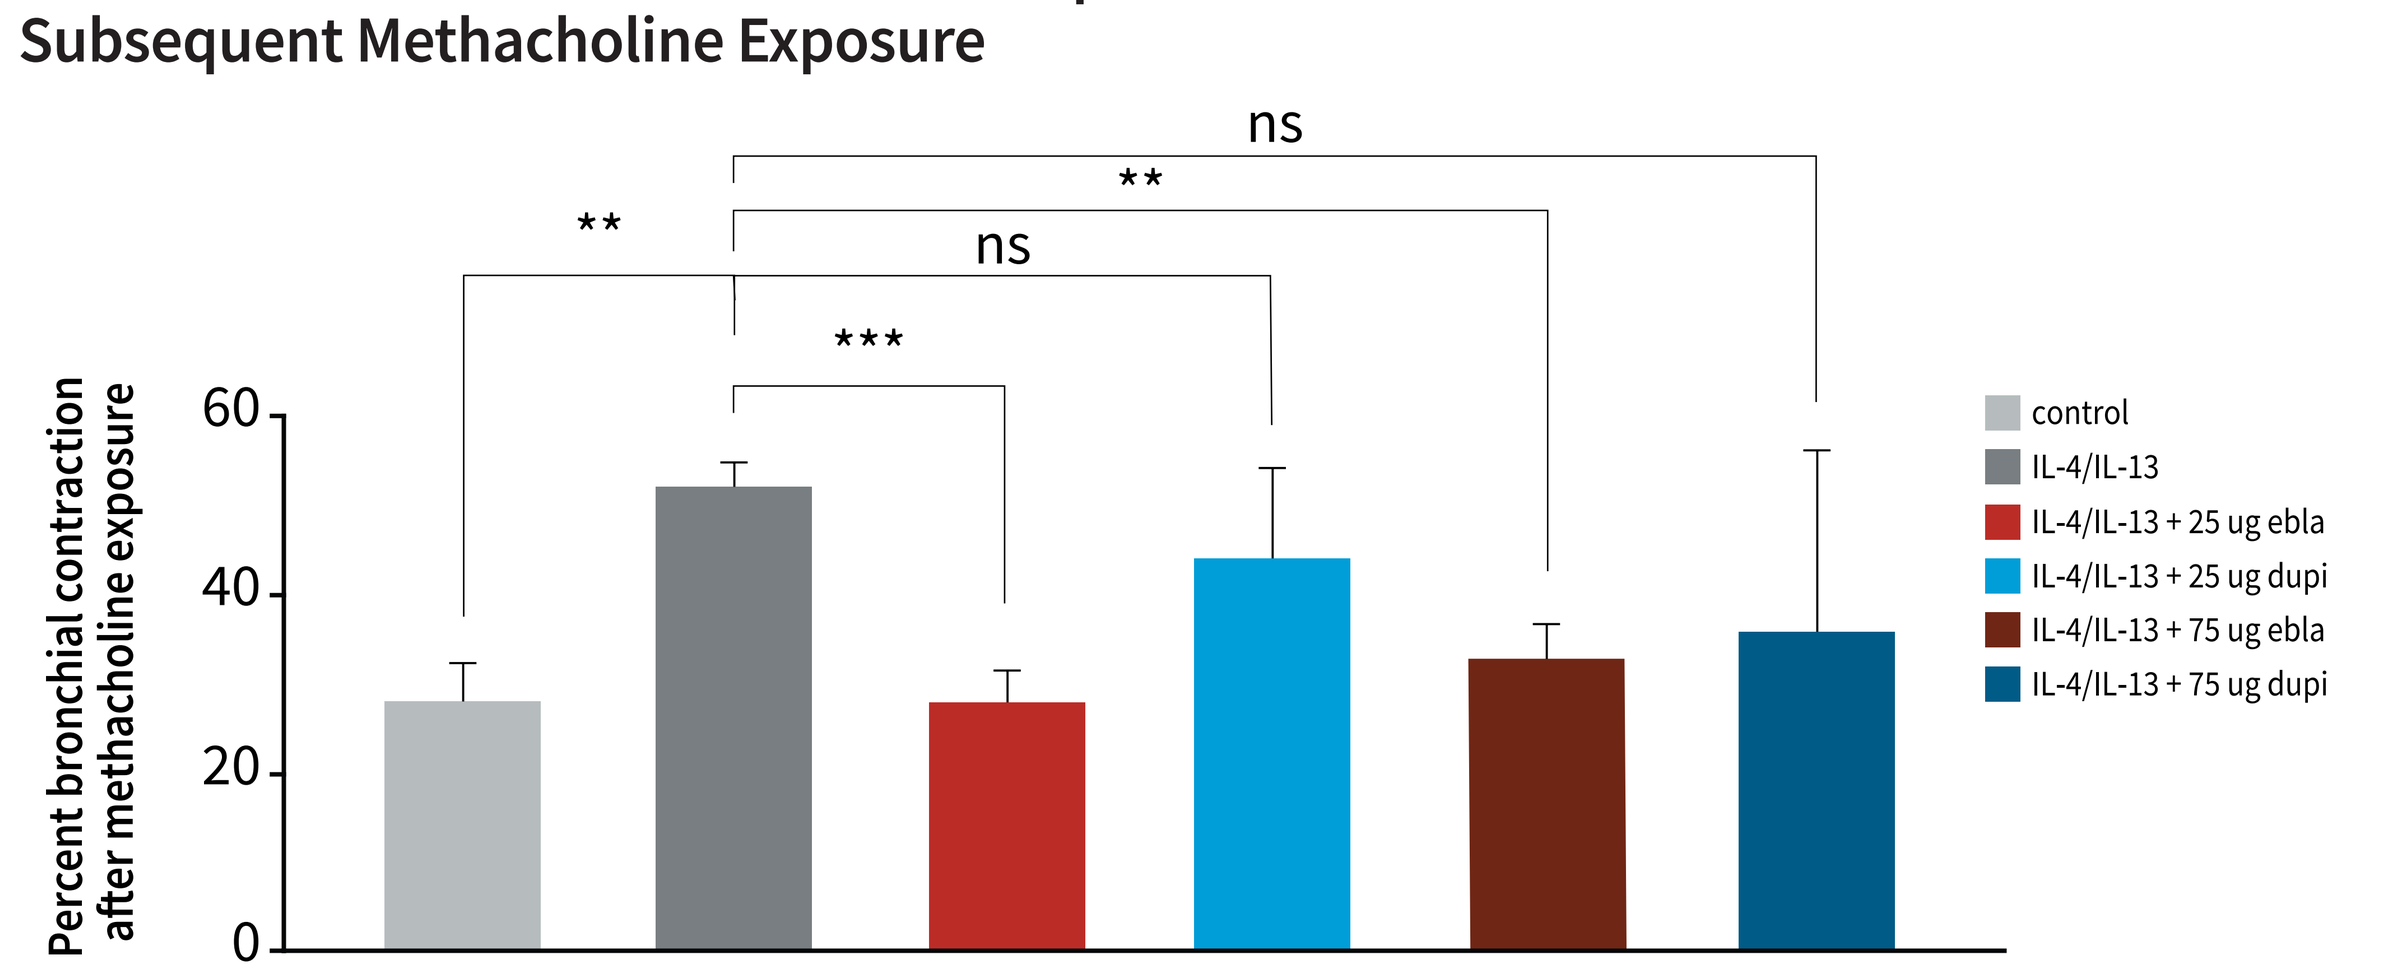
Percent bronchial contraction after methacholine exposure
60
40
20
0
ns
**
**
ns

control
IL-4/IL-13
IL-4/IL-13 + 25 ug ebla
IL-4/IL-13 + 25 ug dupi
IL-4/IL-13 + 75 ug ebla
IL-4/IL-13 + 75 ug dupi

COPD, chronic obstructive pulmonary disease; dupi, dupilumab; ebla, eblasakimab; ns, not significant.
n=6-13 airways per condition.
p*<0.05; *p*<0.01; ****p*<0.001.

METHODS

Translational Lung Model

- Precision-cut lung slices (PCLS) from a 66-year-old male with no known history of lung diseases and a 67-year-old female with COPD were used to test constriction and dilation responses in response to different treatment conditions
- PCLS were assigned to the following treatment groups, each administered for 48 hours: untreated control, cytokine-stimulated with combination IL-4 and IL-13 treatment (250ng/mL IL-4 + 250ng/ml IL-13), or cytokine-stimulated and co-treated with different concentrations of eblasakimab or dupilumab as shown in the graphs (either 25 µg/mL, 75 µg/mL, or 500 µg/mL doses) (**Figure 2**)
 - Drug concentrations were selected to be within range of steady-state serum concentrations of dupilumab;[9] concentrations were used to develop a head-to-head eblasakimab vs dupilumab study design
 - A minimum of 6 PCLS were tested for each treatment condition
- PCLS were individually placed in separate wells of 6-well plates grouped by the experimental treatment conditions described above; the multi-well plates were mounted on a computer-controlled translation stage and imaged using an inverted microscope (DMI6000B; Leica Microsystems)
- Following 48-hour treatment, PCLS were sequentially treated with 10nM of the broncho-dilating agent formoterol, briefly rinsed with conditioned medium (without formoterol), then treated with 10µM of the constricting agent methacholine
- PCLS were imaged at baseline, 5 and 15 minutes after formoterol treatment, and 20 minutes after methacholine treatment
- From the acquired images, airway luminal area was quantified (Image J software; National Institutes of Health, Bethesda, MD) and its magnitude normalized to 15 minutes of formoterol treatment

FIGURE 2. SCHEMATIC OF TRANSLATIONAL LUNG MODEL

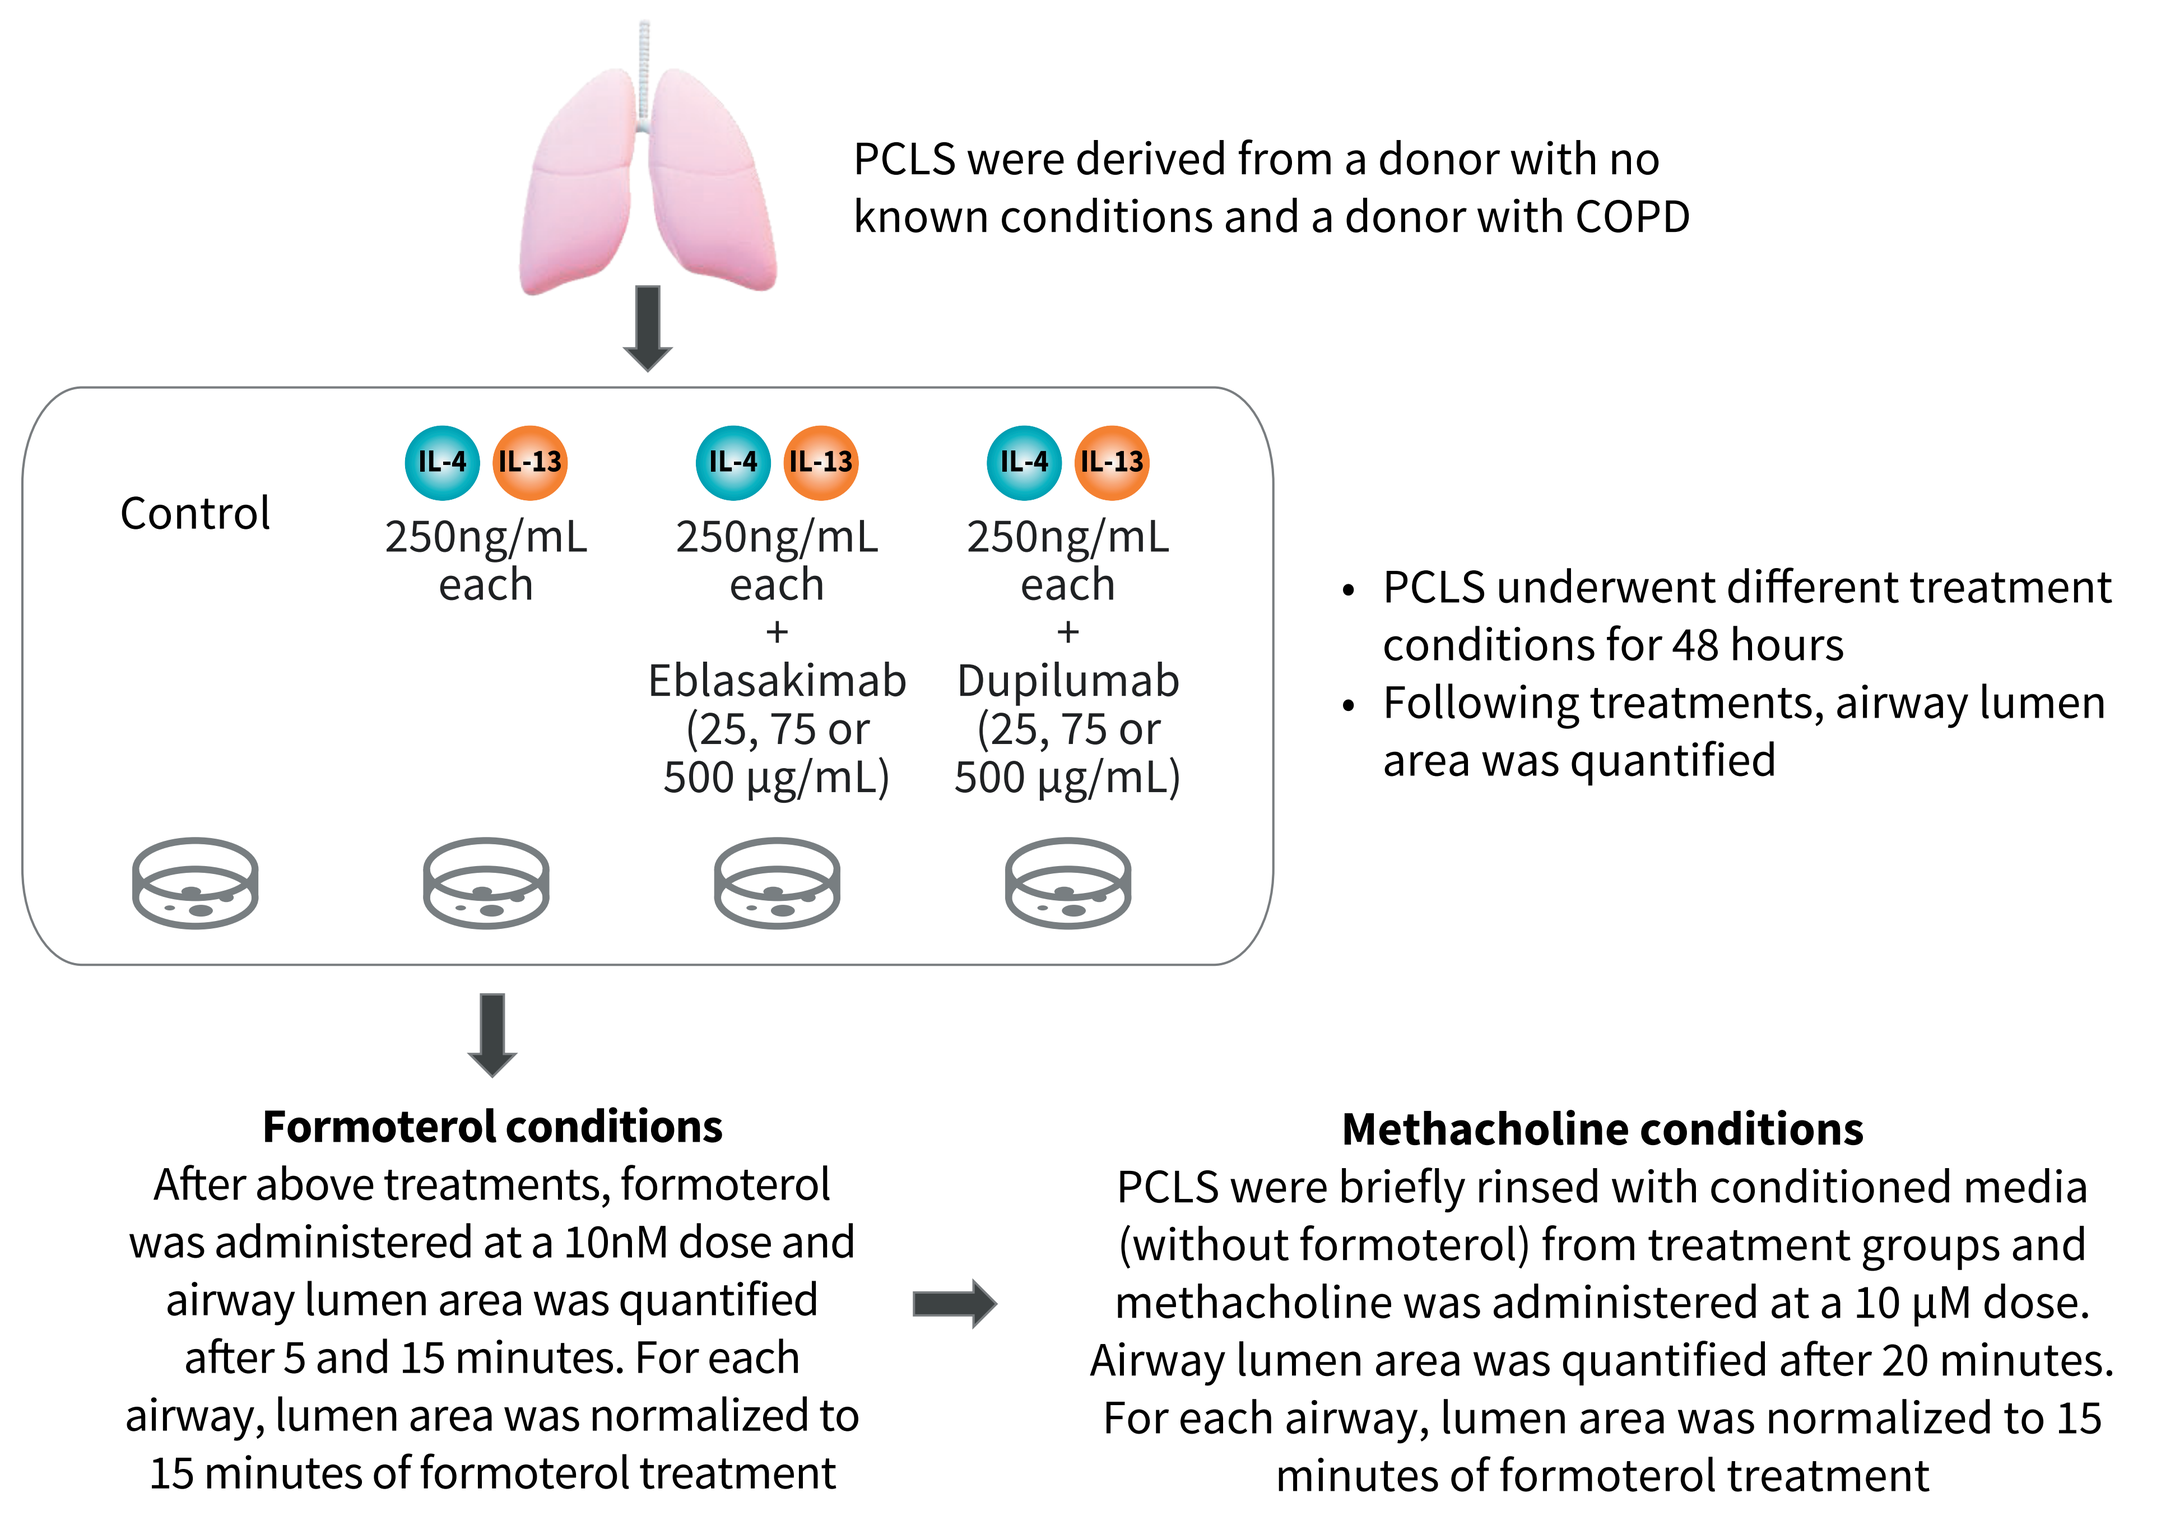
PCLS were derived from a donor with no known conditions and a donor with COPD
Control
IL-4 IL-13
250ng/mL each
250ng/mL each + Eblasakimab (25, 75 or 500 µg/mL)
250ng/mL each + Dupilumab (25, 75 or 500 µg/mL)
• PCLS underwent different treatment conditions for 48 hours
• Following treatments, airway lumen area was quantified
Formoterol conditions
After above treatments, formoterol was administered at a 10nM dose and airway lumen area was quantified after 5 and 15 minutes. For each airway, lumen area was normalized to 15 minutes of formoterol treatment
Methacholine conditions
PCLS were briefly rinsed with conditioned media (without formoterol) from treatment groups and methacholine was administered at a 10 µM dose. Airway lumen area was quantified after 20 minutes. For each airway, lumen area was normalized to 15 minutes of formoterol treatment

COPD, chronic obstructive lung disease; IL-4, interleukin-4; IL-13, interleukin-13; PCLS, precision-cut lung slices.

Statistical Analysis

- Unpaired t-tests assuming parametric distribution were performed to compare treatment groups, with p<0.05 considered statistically significant.

CONCLUSIONS

- In both healthy and COPD-derived lung tissues, eblasakimab significantly reduced IL-4- and IL-13-induced bronchial airway constriction, both pre- and post- methacholine stimulation
- In IL-4 and IL-13 pre-treated healthy and COPD-derived lung tissues, eblasakimab further restored formoterol-induced airway dilation and improved methacholine-induced constriction
- Results suggest a therapeutic benefit of eblasakimab in reducing IL-4- and IL-13-induced airway hyperresponsiveness and that eblasakimab may provide relief in situations of acute airway constriction
- Eblasakimab showed significant improvement across all measured bronchial outcomes whereas dupilumab did not achieve statistical significance relative to placebo for all measures, suggesting that eblasakimab may potentially provide stronger relief against bronchoconstriction as well as improved dilatory function in COPD compared to dupilumab
 - These results may be attributed to the potential of eblasakimab to inhibit the Type II receptor complex more efficiently, resulting in greater reduction in Th2 cytokine activity
- As airway hyperresponsiveness is clinically shared across asthma, COPD, and other lung disorders, eblasakimab has the potential to improve airway function in various Th2-driven lung conditions
- Taken together, these preliminary results support further investigation of eblasakimab as a potential therapeutic option for COPD with potentially more effective blockade of Type-2 mediated effects in lung tissue

REFERENCES

1. Celli BR, et al. *Am J Respir Crit Care Med.* 2023;207(9):1134-1144.
2. Patalano F, et al. *Eur Respir Rev.* 2014;23(133):333-44.
3. Oishi K, et al. *J Clin Med.* 2020;9(8):2670.
4. Bhatt et al. *N Engl J Med.* 2023;389(3):205-214.
5. Bao K, Reinhardt RL. *Cytokine.* 2015;75(1):25-37.
6. Junttila IS. *Front Immunol* 2018; 9:888.
7. Le Floc'h A, et al. *Allergy.* 2020;75:1188-1204.
8. Cevikbas F, et al. *Clin Immunol.* 2023;253:109677.
9. Center for Drug Evaluation and Research. Application number: 761055orig1s000. Clinical Pharmacology and Biopharmaceutics review(s). Accessed April 8, 2024. https://www.accessdata.fda.gov/drugsatfda_docs/nda/2017/761055Orig1s000ClinPharmR.pdf

ACKNOWLEDGEMENTS

ASLAN Pharmaceuticals thanks Mechanobiologix for conducting the studies and Dr. Ramaswamy Krishnan for his expert advice. Dani Guralnick provided graphic design support in developing the eblasakimab and dupilumab schematics. Medical writing support was provided by Prescott Medical Communications Group, a Citrus Health Group, Inc. company (Chicago, IL), and was funded by ASLAN Pharmaceuticals.

AUTHOR DISCLOSURES

This study was funded by ASLAN Pharmaceuticals. FC and CF are employees of ASLAN Pharmaceuticals.

Poster presented at the American Thoracic Society 2024 | San Diego, CA | May 17-22, 2024