Abstract Number: LB793

A phase 1, open-label, single ascending dose study in healthy subjects of the safety, tolerability and pharmacokinetics of ASLAN004, a novel IgG anti-IL-13 receptor alpha 1 Inhibitor

Lawrence Soon-U Lee,[1] Hartina Hajireen,[2] Alison Ward,[3] Carl Firth[3]

1. National University of Singapore, Singapore 2. Clinical Trials & Research Unit, Changi General Hospital, Singapore. 3. ASLAN Pharmaceuticals Pte Ltd, Singapore.

Abstract

ASLAN004 is a novel fully human IgG4 anti-IL-13 receptor alpha 1 (IL-13Rα1) monoclonal antibody, that blocks the signaling of IL-4 and IL-13 through the Type II receptor, hence is a potential therapy for atopic dermatitis, asthma and diseases of related etiology. The study aim was to evaluate the safety, tolerability and pharmacokinetics of single ascending doses of ASLAN004 in healthy male subjects. The study had 2 parts: in the first, 5 sequential ascending dose cohorts (0.1 to 10 mg/Kg) were administered ASLAN004 intravenously as a single dose. In the second, 4 cohorts (75 to 600 mg) were administered ASLAN004 subcutaneously, as a single dose. The primary outcome measure was safety and tolerability; secondary outcomes comprised of pharmacokinetic parameters. Pharmacodynamic outcomes were assessed as exploratory endpoints and included IL-13Rα1 receptor occupancy and the inhibition of STAT6 phosphorylation. Peak ASLAN004 serum concentration was achieved 2–4 hours post intravenous administration and approximately 4 days post subcutaneous administration. Duration of pharmacodynamic effect was shown to increase with increasing ASLAN004 intravenous and subcutaneous dose. In all cases, subcutaneous administration showed a higher degree of subject variation for both pharmacokinetic and pharmacodynamic effect when compared with intravenous administration. As a single dose, ASLAN004 was well tolerated with no adverse events that led to study discontinuation and no serious adverse events were reported. Mild itch at the injection site was reported in one individual, resolving within 24 hours. There were no adverse events of special interest associated with ASLAN004. Data from this study support and encourage further development of ASLAN004. Clinical trials registration: NCT03721263.

Introduction

ASLAN004 is a fully human recombinant monoclonal IgG4 antibody that binds specifically to IL-13Rα1 which together with IL-4Rα forms the Type II receptor complex (Figure 1). By binding to the Type II receptor complex, ASLAN004 inhibits the signaling of both IL-4 and IL-13. IL-4 and IL-13 are Th2 inflammatory cytokines which are important to atopic/allergic diseases, such as atopic dermatitis and asthma.

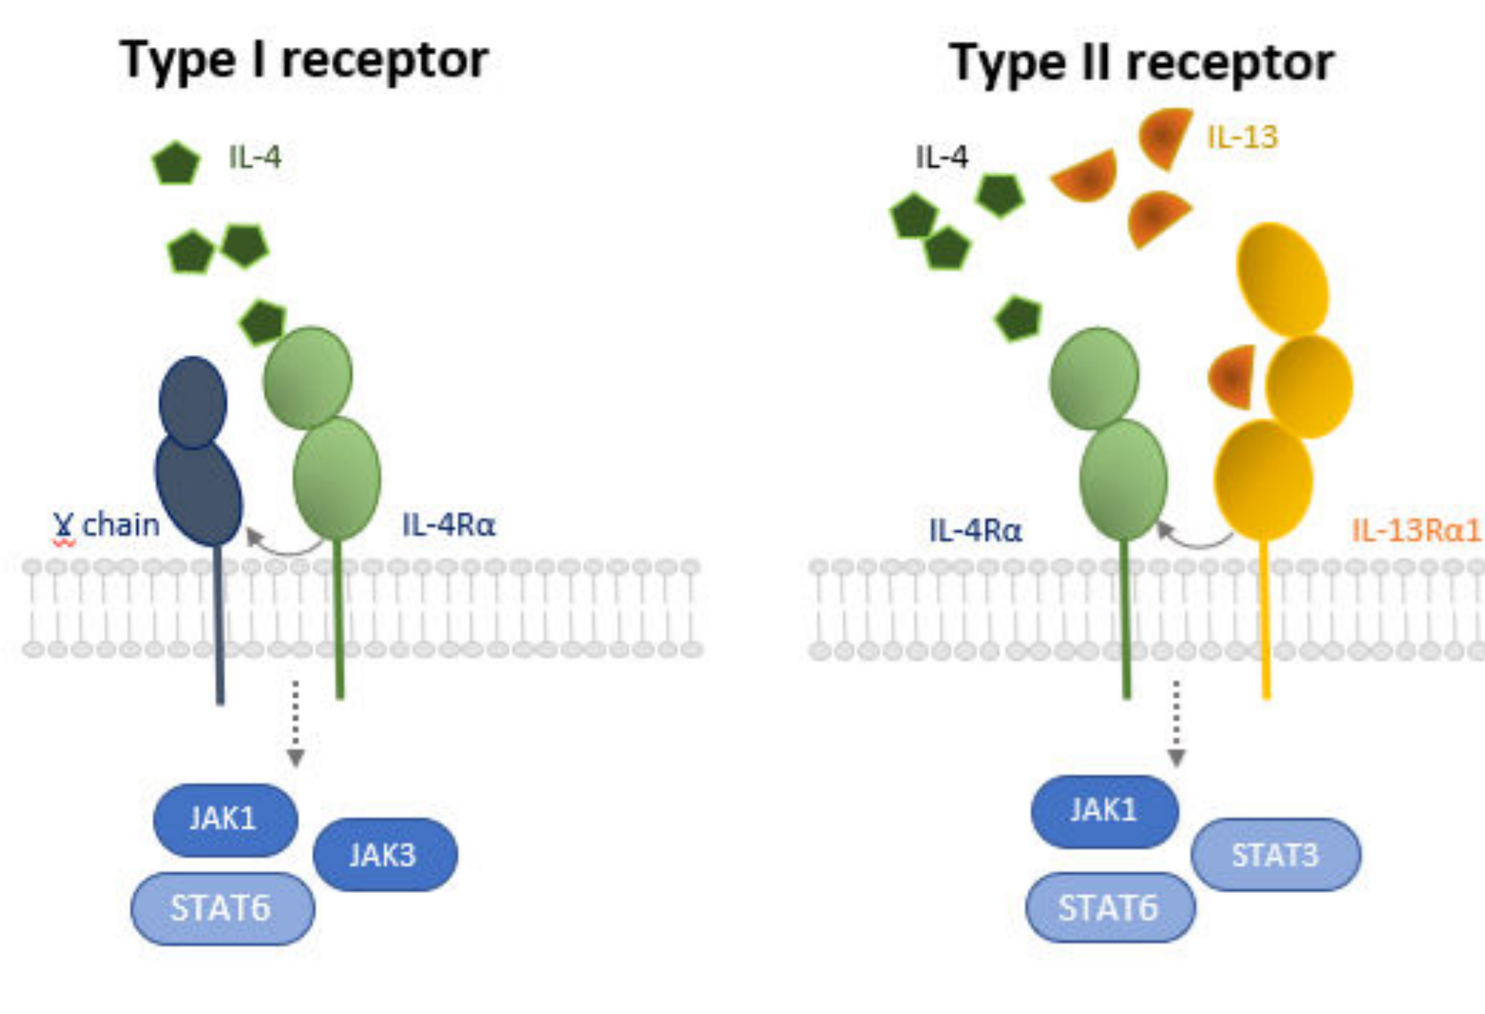
Type I receptor
IL-4
γ chain
IL-4Rα
JAK1
JAK3
STAT6
Type II receptor
IL-4
IL-13
IL-4Rα
IL-13Rα1
JAK1
STAT3
STAT6

Figure 1: Type I and II receptor complexes

ASLAN004-001 was an open-label, two-part, single-center, first-in-human, single ascending dose (SAD) study to assess the effects of single doses of ASLAN004 when administered to healthy male subjects. The study objective was to evaluate safety, tolerability, and pharmacokinetics (PK) in healthy male subjects to support future development in inflammatory or allergic disorders, including atopic dermatitis. Pharmacodynamic (PD) outcomes were assessed as exploratory endpoints and included IL-13Rα1 receptor occupancy and the inhibition of STAT6 phosphorylation.

Methodology

In the SAD, ASLAN004 was administered intravenously (IV) and subcutaneously (SC) in 9 cohorts of subjects, with a total of 44 subjects being dosed with ASLAN004. The cohorts in the study received ASLAN004 as follows: 0.1 mg/Kg IV (N=2), 0.3 mg/Kg IV (N=3), 1 mg/Kg IV (N=3), 3 mg/Kg IV (N=6), 10 mg/Kg IV (N=6), 75 mg SC (N=6), 150 mg SC (N=6), 300 mg SC (N=6), and 600 mg SC (N=6) (Figure 2).

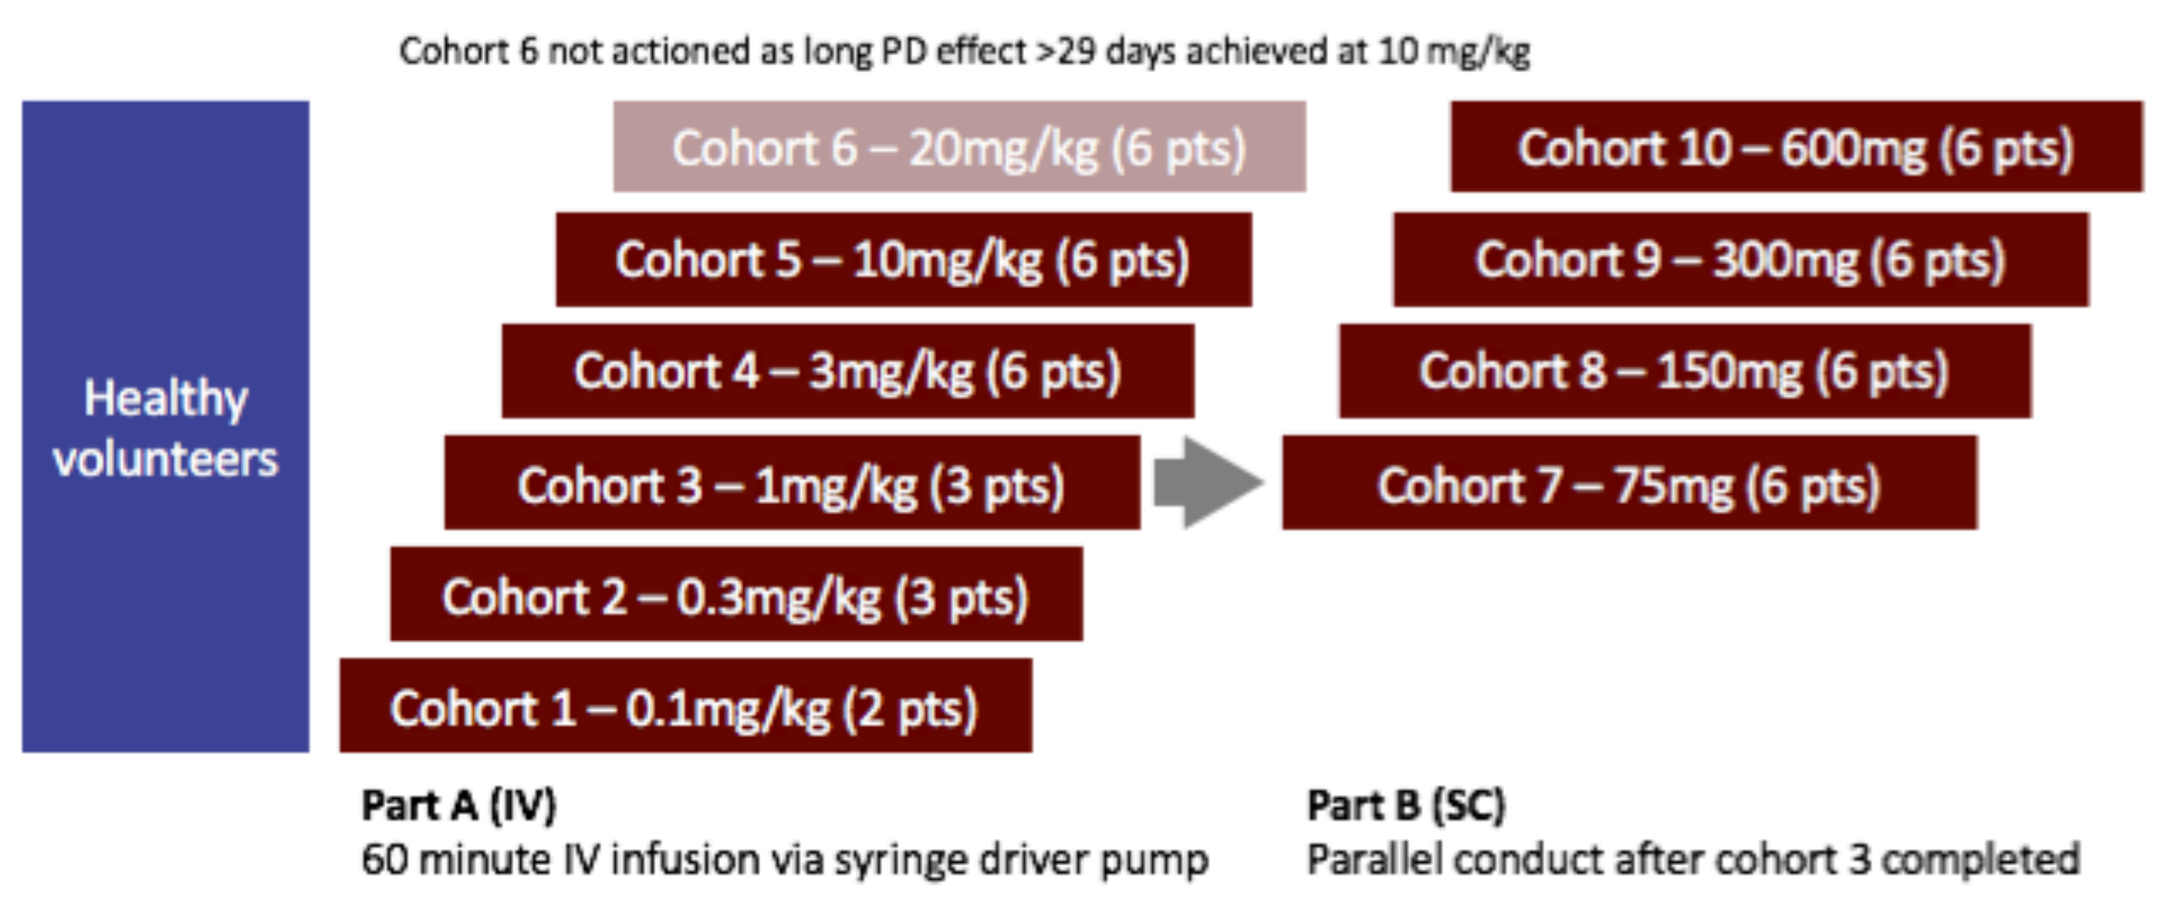
Cohort 6 not actioned as long PD effect >29 days achieved at 10 mg/kg
Healthy volunteers
Cohort 6 – 20mg/kg (6 pts)
Cohort 5 – 10mg/kg (6 pts)
Cohort 4 – 3mg/kg (6 pts)
Cohort 3 – 1mg/kg (3 pts)
Cohort 2 – 0.3mg/kg (3 pts)
Cohort 1 – 0.1mg/kg (2 pts)
Cohort 10 – 600mg (6 pts)
Cohort 9 – 300mg (6 pts)
Cohort 8 – 150mg (6 pts)
Cohort 7 – 75mg (6 pts)
Part A (IV)
60 minute IV infusion via syringe driver pump
Part B (SC)
Parallel conduct after cohort 3 completed

Figure 2: ASLAN004-001 Study Design

The sponsor-initiated escalation to the next sequential dosing cohort, after safety data reviews by the Safety Monitoring Committee (SMC), confirmed that no safety signals were identified and no study stopping rules were met. The IV cohorts were initiated first. The first SC cohort, 75 mg, was started once the 1 mg/Kg IV cohort had completed (based on the assumption that an average healthy subject weight was 75 Kg and given anticipated higher drug exposure via the IV route). Subsequently, the IV and SC cohorts ran independently. After dose administration of each SAD SC cohort was completed, there was a safety monitoring review by the SMC of the 7 days post-dose safety data to assess the subjects and the decision was made to proceed with the next dose.

Safety Assessments

Safety assessments included vital signs, AEs, ECG, clinical laboratory tests (hematology, coagulation, clinical chemistry, urinalysis, and serology) and physical examinations.

Immunogenicity

Anti-drug antibodies were measured.

Pharmacokinetic (PK) Assessments

ASLAN004 quantified in serum was measured using a validated immunoassay. In the assay, ASLAN004 binds to IL-13Rα1 coated microtiter plates and is then detected using a sulfo-labelled anti-idiotype human antibody (hu 1B5-G4pk-a10G5-6) and light emission following addition of ECL reagent. Serum samples were collected for ASLAN004 PK analysis at pre-dose, 1, 2, 4 and 8 hr and days 2, 4, 8, 15, 22, 29, 26, 43, 57 and 85 post dose.

Pharmacodynamic (PD) Assessments

Assessments were made at pre-dose and 1 hr, 24 hr, days 8, 15, 29 and 85 post dose using whole blood assays. IL-13Rα1 receptor occupancy (RO) and inhibition of STAT6 phosphorylation were used as pharmacodynamic markers. Pharmacodynamic assessments were assessed using a number of immune cells, including monocytes, lymphocytes and B-cells.

Results

Safety Assessments

At the study conclusion, 16 out of 44 subjects had experienced at least one Treatment-Emergent Adverse Event (TEAE) regardless of causality. Five out of 44 subjects experienced at least one related TEAE. The incidence of the most frequent TEAEs when ASLAN004 was administered by the IV route only was: upper respiratory tract infection (20%), decreased appetite (15%), headache (15%), oropharyngeal pain (15%) and pyrexia (15%). The combined incidence of the most frequent TEAEs when ASLAN004 was administered by IV or SC route was upper respiratory tract infection (11.4%) and headache (11.4%). Mild itch at the injection site was reported in one individual, resolving within 24 hours. A full list of TEAEs is presented in Table 1 by route of administration. None of the subjects experienced any serious adverse event or were withdrawn from the study. There was no dose response for TEAEs. There were no fatalities during the study. No safety signals were observed.

Preferred term	Number subject (%) IV (N=20)	Number of events IV (N=20)	Number subjects (%) SC (N=24)	Number of events SC (N=24)	TOTAL subjects (%) IV & SC (N=44)	TOTAL number of events IV & SC (N=44)
No. of subjects with at least one TEAE	10 (50.0%)	38	8 (33.3%)	14	18 (40.9%)	52
Infections & Infestations	5 (25%)	5	2 (8.3%)	2	7 (15.9%)	7
Upper respiratory tract infection	4 (20.0%)	5	1 (4.2%)	1	5 (11.4%)	6
Pharyngitis	1 (5.0%)	1	0	0	1 (2.3%)	1
Gastroenteritis	0	0	1 (4.2%)	1	1 (2.3%)	1
Investigations	5 (25.0%)	12	2 (8.3%)	4	7 (15.9%)	16
C-reactive protein increased	2 (10.0%)	2	2 (8.3%)	2	4 (9.1%)	4
Monocyte count increased	2 (10.0%)	2	0	0	2 (4.5%)	2
Neutrophil count increased	2 (10.0%)	2	0	0	2 (4.5%)	2
White blood cell count increased	2 (10.0%)	2	0	0	2 (4.5%)	2
Alanine aminotransferase increased	1 (5.0%)	1	0	0	1 (2.3%)	1
Neutrophil count decreased	1 (5.0%)	1	0	0	1 (2.3%)	1
Weight decreased	1 (5.0%)	1	0	0	1 (2.3%)	1
White blood cell count decreased	1 (5.0%)	1	0	0	1 (2.3%)	1
Blood lactate dehydrogenase increased	0	0	1 (4.2%)	1	1 (2.3%)	1
Lymphocyte count decreased	0	0	1 (4.2%)	1	1 (2.3%)	1
Nervous system disorders	4 (20.0%)	4	2 (8.3%)	2	6 (13.6%)	6
Headache	3 (15.0%)	3	2 (8.3%)	2	5 (11.4%)	5
Lethargy	1 (5.0%)	1	0	0	1 (2.3%)	1
General disorders & administration site conditions	3 (5.0%)	4	3 (12.5%)	3	6 (13.6%)	7
Pyrexia	3 (15.0%)	3	1 (4.2%)	1	4 (9.1%)	4
Fatigue	1 (5.0%)	1	0	0	1 (2.3%)	1
Injection site pruritis	0	0	1 (4.2%)	1	1 (2.3%)	1
Non cardiac chest pain	0	0	1 (4.2%)	1	1 (2.3%)	1
Metabolism & nutrition disorders	3 (15.0%)	3	1 (4.2%)	1	4 (9.1%)	4
Decreased appetite	3 (15.0%)	3	1 (4.2%)	1	4 (9.1%)	4
Respiratory, thoracic & mediastinal disorders	3 (15.0%)	6	0	0	3 (6.8%)	6
Oropharyngeal pain	3 (15.0%)	3	0	0	3 (6.8%)	3
Cough	1 (5.0%)	1	0	0	3 (6.8%)	3
Dysphonia	1 (5.0%)	1	0	0	1 (2.3%)	3
Rhinorrhoea	1 (5.0%)	1	0	0	1 (2.3%)	3
Musculoskeletal & connective tissue disorders	2 (10.0%)	2	0	0	2 (4.5%)	2
Muscle tightness	1 (5.0%)	1	0	0	1 (2.3%)	1
Musculoskeletal pain	1 (5.0%)	1	0	0	1 (2.3%)	1
Ear & labyrinth disorders	1 (5.0%)	1	0	0	1 (2.3%)	1
Ear discomfort	1 (5.0%)	1	0	0	1 (2.3%)	1
Gastrointestinal disorders	1 (5.0%)	1	2 (8.3%)	2	3 (6.8%)	3
Abdominal distension	1 (5.0%)	1	0	0	1 (2.3%)	1
Diarrhoea	0	0	1 (4.2%)	1	1 (2.3%)	1
Toothache	0	0	1 (4.2%)	1	1 (2.3%)	1

Table 1. Incidence of TEAE Related to ASLAN004

Pharmacokinetic (PK) Assessments

There was a rapid increase in exposure to ASLAN004 when given IV, with maximal concentration achieved around 1 to 3 hours post dose (median) (Figure 3).

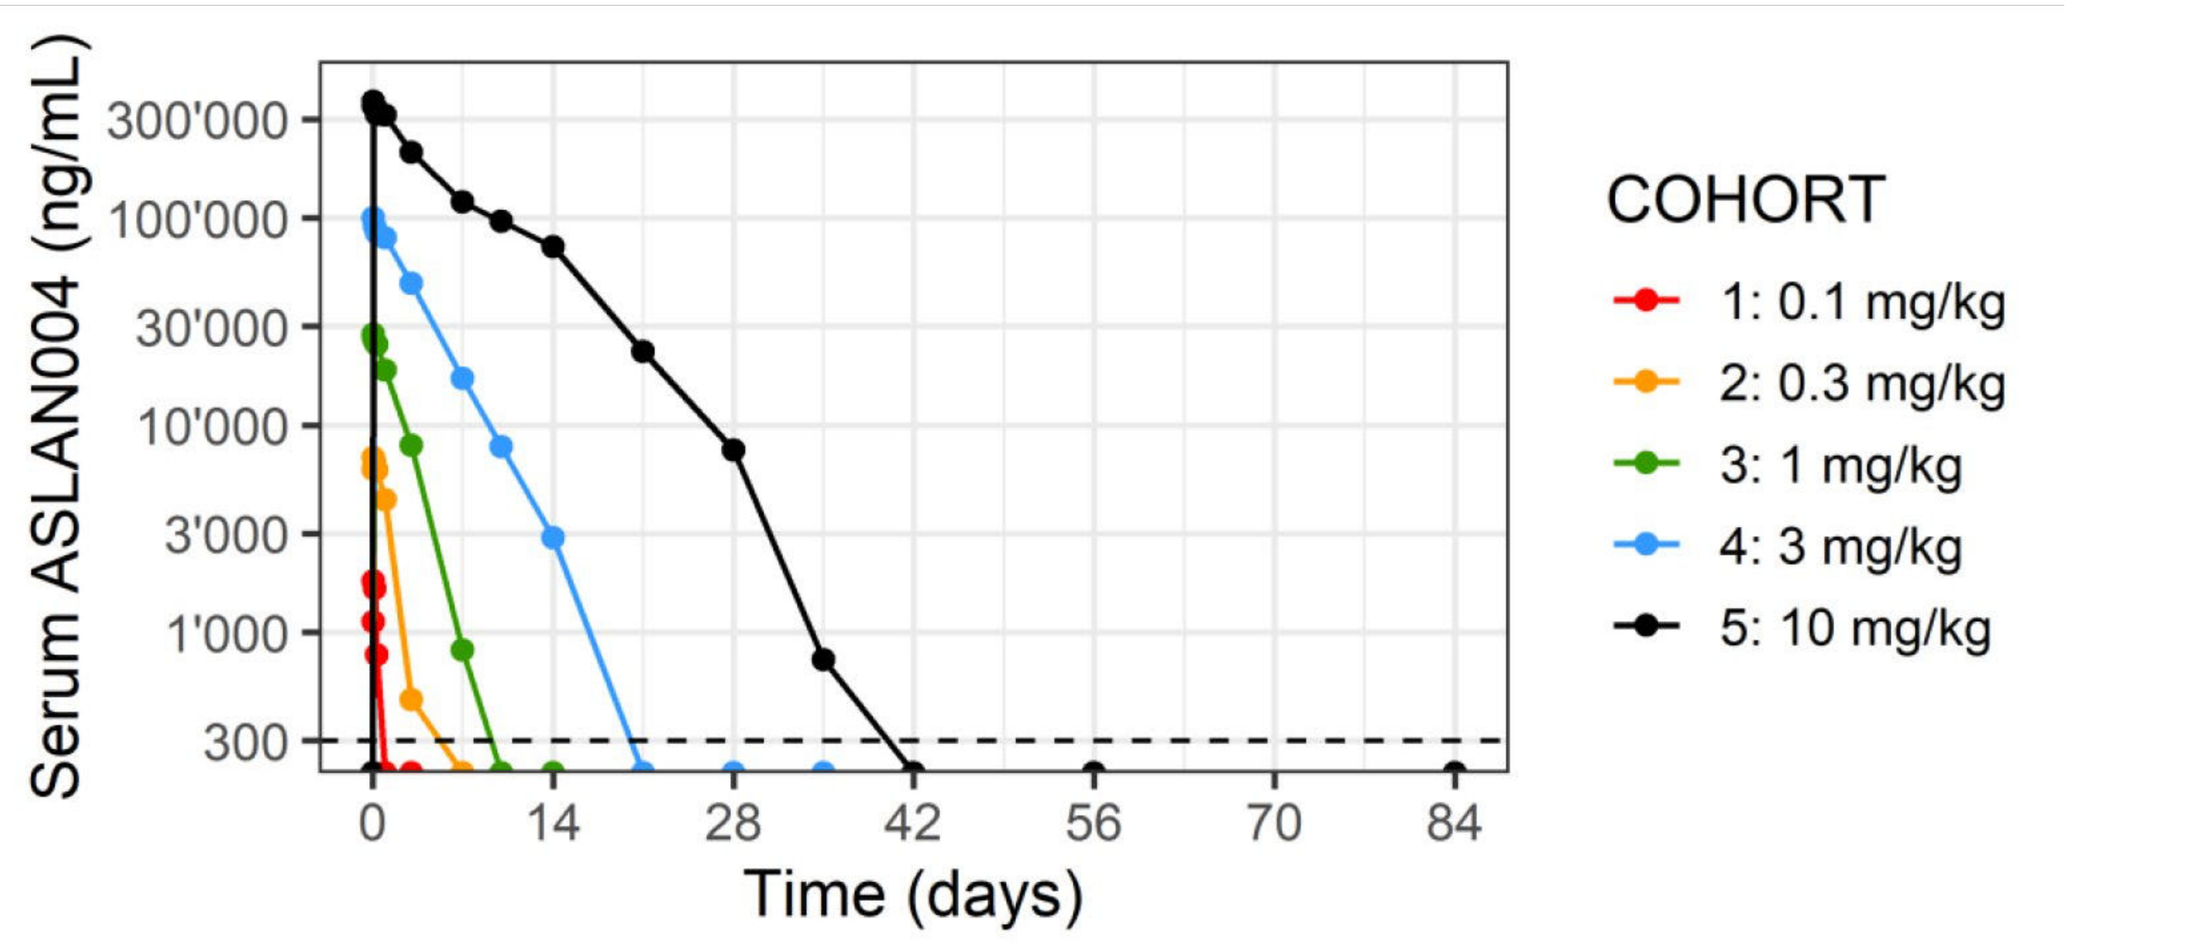
Serum ASLAN004 (ng/mL)
300'000
100'000
30'000
10'000
3'000
1'000
300
0
14
28
42
56
70
84
Time (days)
COHORT
1: 0.1 mg/kg
2: 0.3 mg/kg
3: 1 mg/kg
4: 3 mg/kg
5: 10 mg/kg

Figure 3: ASLAN004 delivered IV: ASLAN004 serum concentration to time profile on a semi logarithmic scale

As expected, delivery of ASLAN004 SC took a longer period to achieve peak serum concentrations (C_{max}). ASLAN004 serum

SC administration of ASLAN004 gave a lower exposure to drug than the corresponding dose given IV. The maximum dose administered to patients in the 10 mg/kg cohort was 899 mg. Non-dose proportional PK behavior was seen with ASLAN004 administered via both the IV and SC route, in accordance with observations made in earlier Cynomolgus monkey studies. This PK behavior is attributed to target mediated clearance of ASLAN004.

Immunogenicity

No immunogenicity to ASLAN004 was determined in humans.

Pharmacodynamic (PD) Assessments

There was a dose dependent PD effect that followed the pharmacokinetic profile. This was more pronounced in the IV cohorts, where less variability between subjects was observed. At lower IV doses pSTAT6 inhibition was not seen until 1 mg/Kg. At higher IV doses (10 mg/Kg), complete PD effect was observed to at least 29 days. At 300 mg and 600 mg SC, a complete PD effect was observed for at least 15 days.

ASLAN004 PD mean ± SD effect for IL-13Rα1 receptor occupancy and inhibition of pSTAT6 in monocytes are shown for SC doses in Figures 4–7.

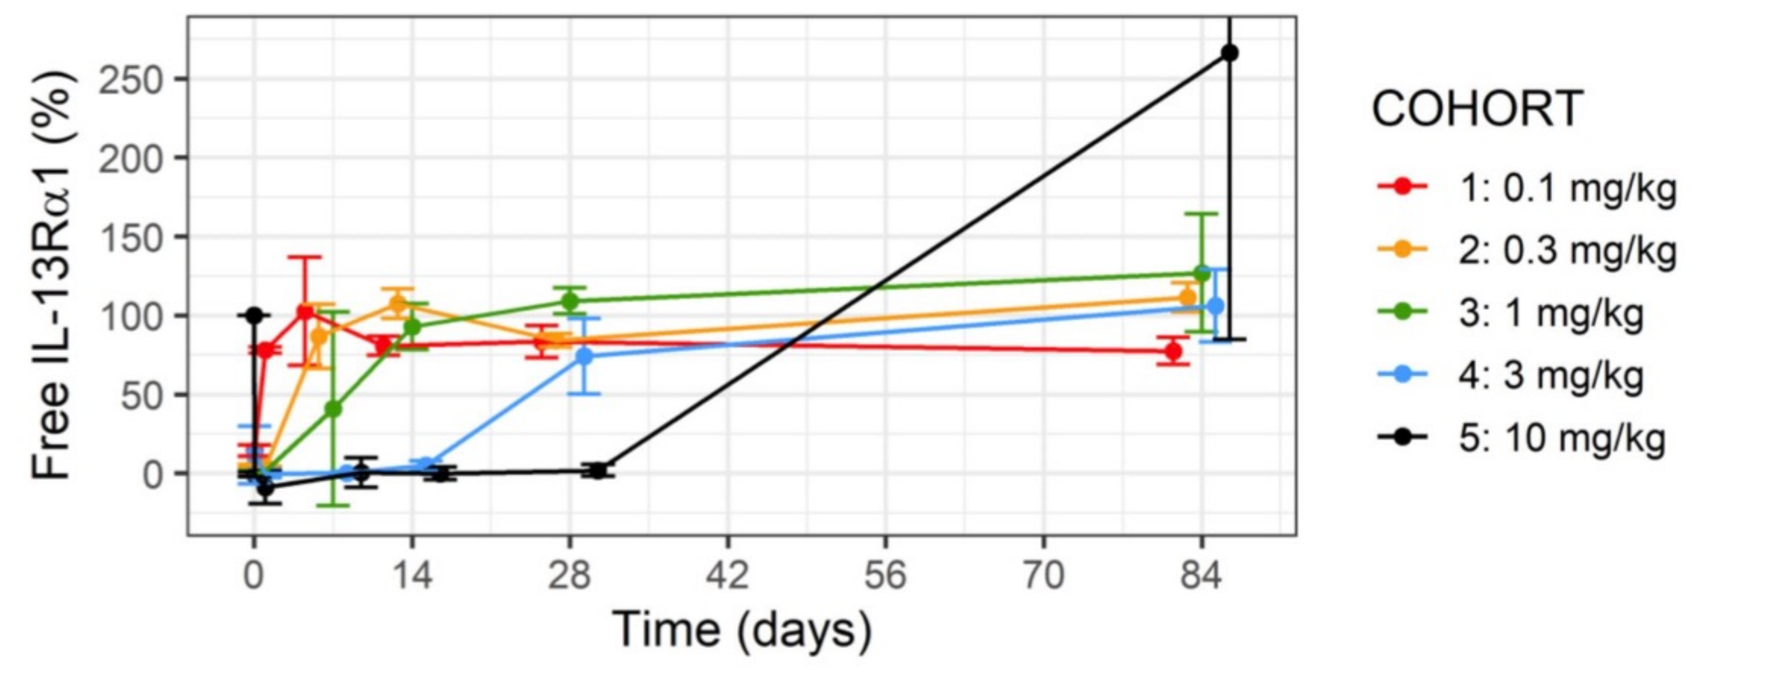
Free IL-13Rα1 (%)
250
200
150
100
50
0
0
14
28
42
56
70
84
Time (days)
COHORT
1: 0.1 mg/kg
2: 0.3 mg/kg
3: 1 mg/kg
4: 3 mg/kg
5: 10 mg/kg

Figure 4: Mean (± SD) IL-13Rα1 receptor occupancy in monocytes for ASLAN004-001 IV cohorts

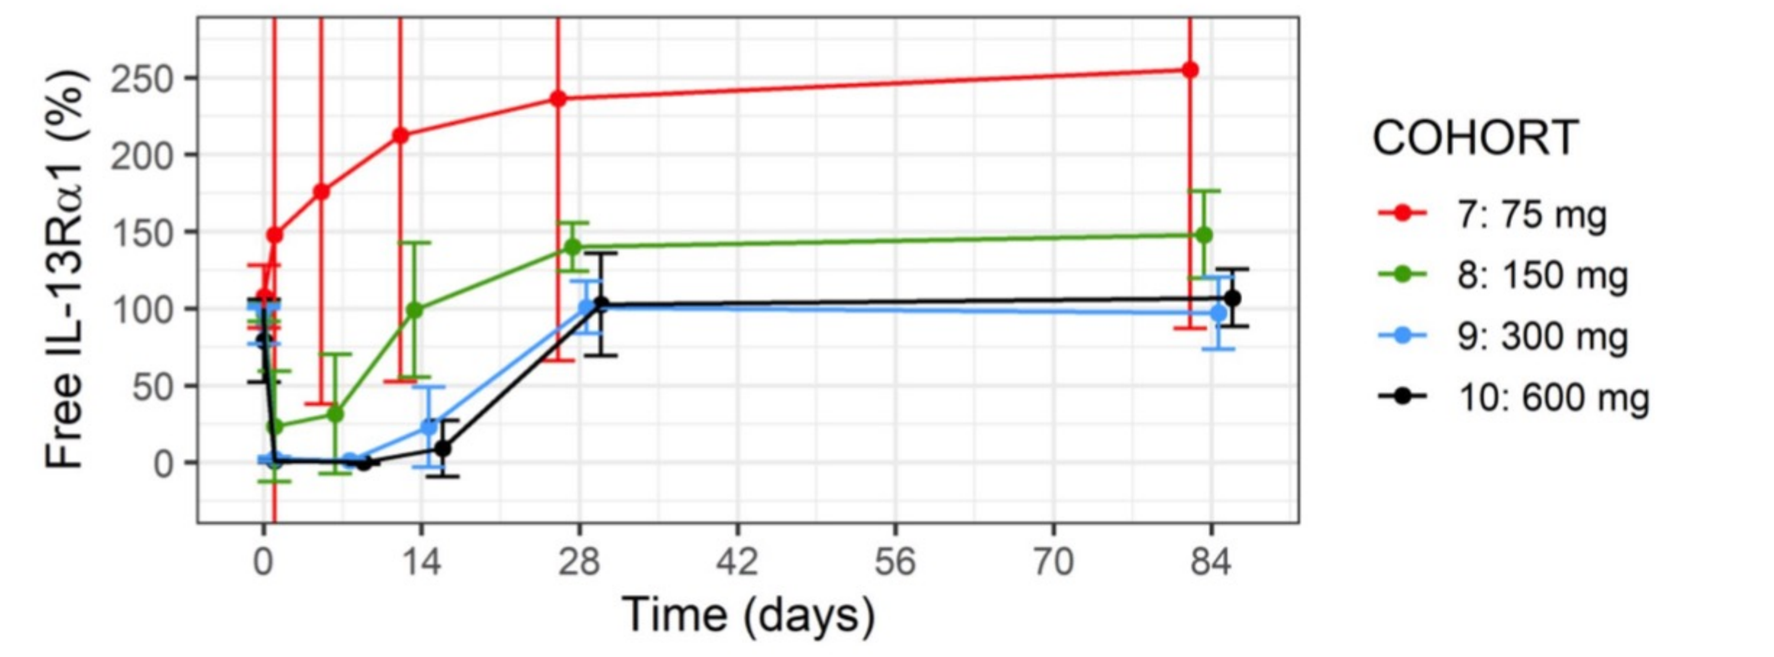
Free IL-13Rα1 (%)
250
200
150
100
50
0
0
14
28
42
56
70
84
Time (days)
COHORT
7: 75 mg
8: 150 mg
9: 300 mg
10: 600 mg

Figure 5: Mean (± SD) IL-13Rα1 receptor occupancy in monocytes for ASLAN004-001 SC cohorts

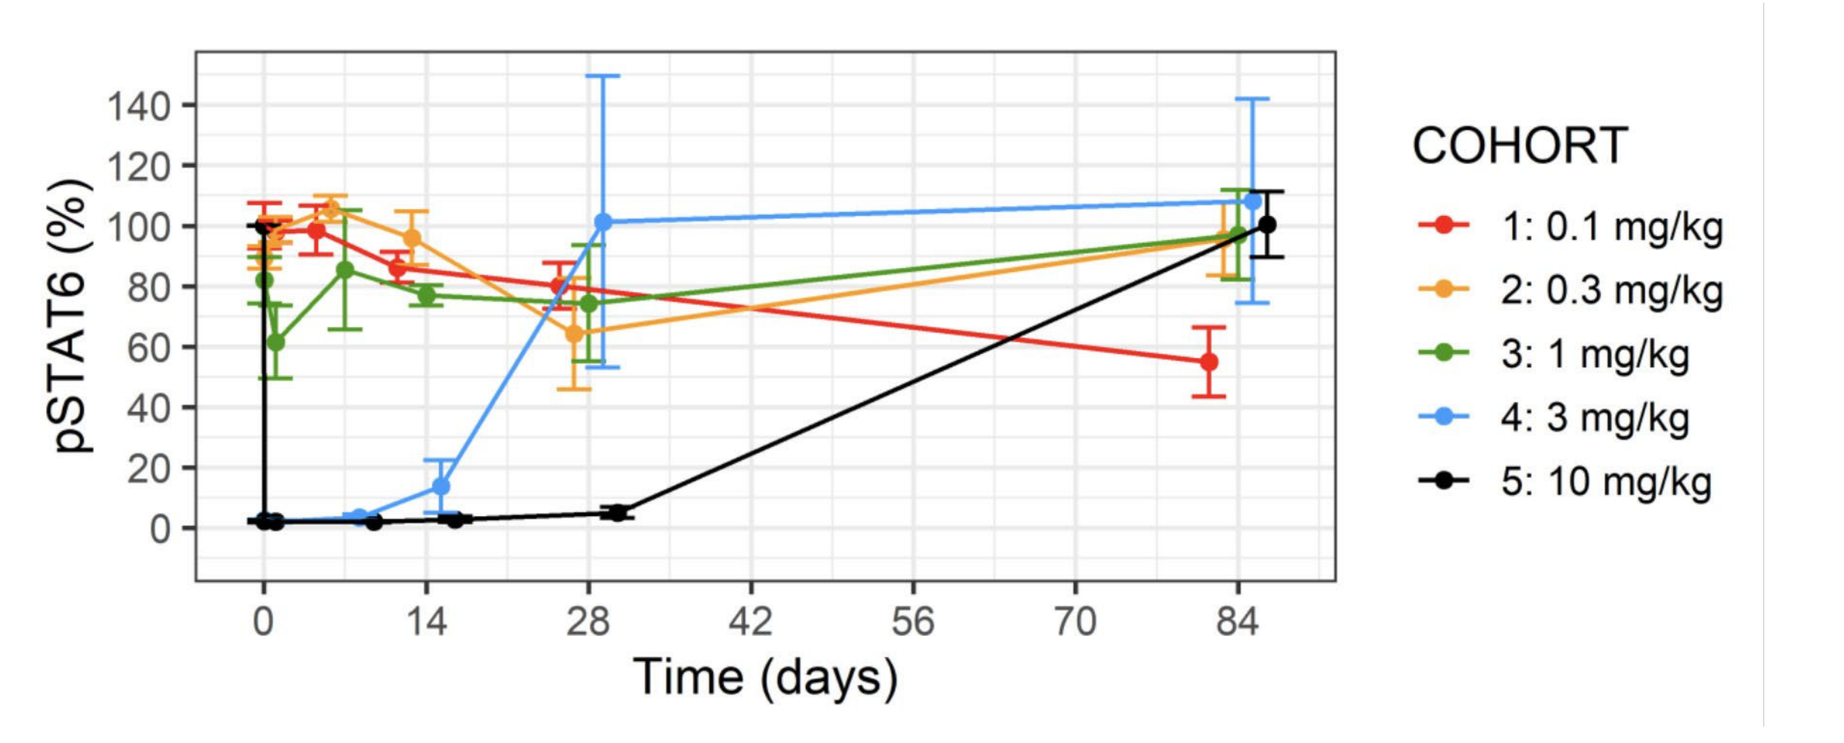
pSTAT6 (%)
140
120
100
80
60
40
20
0
0
14
28
42
56
70
84
Time (days)
COHORT
1: 0.1 mg/kg
2: 0.3 mg/kg
3: 1 mg/kg
4: 3 mg/kg
5: 10 mg/kg

Figure 6: Mean (± SD) inhibition of pSTAT6 in monocytes for ASLAN004- 001 IV cohorts

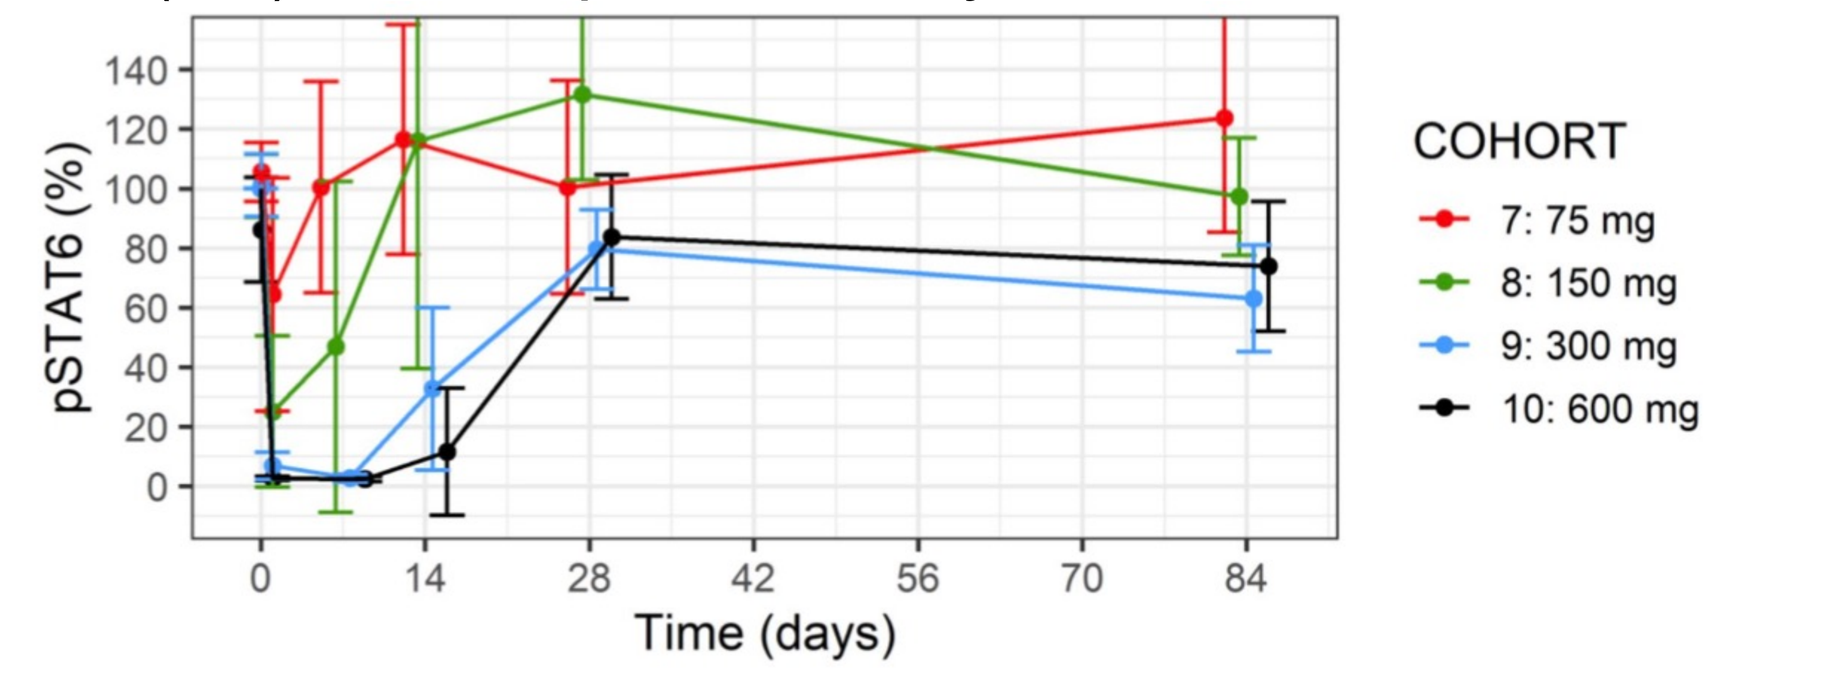
pSTAT6 (%)
140
120
100
80
60
40
20
0
0
14
28
42
56
70
84
Time (days)
COHORT
7: 75 mg
8: 150 mg
9: 300 mg
10: 600 mg

Figure 7: Mean (± SD) inhibition of pSTAT6 in monocytes for ASLAN004- 001 IV cohorts

Conclusion

This study has helped elucidate the pharmacokinetic and pharmacodynamic profile of ASLAN004 and has demonstrated that ASLAN004 is well tolerated with no adverse events that led to study discontinuation. These data provide a strong foundation to advance to the next phase of the clinical trial program. These data are an encouraging first step in exploring ASLAN004’s potential as a novel therapeutic agent targeting the IL-13 receptor with a differentiated approach to treating atopic dermatitis, asthma and diseases of IL-13 related etiology.

Acknowledgments

ASLAN Pharmaceuticals Pte Ltd was the sponsor of this study.